The CBL-B Inhibitor, NX-0255, Improves Human Drug Enhanced Tumor Infiltrating Lymphocyte (DeTIL-0255) Expansion and T-cell Function in Full-scale Runs

Xiaoyan Liang, Xianzhu Wu, Jeevitha Jeevan, Samuel Butler, Pranav Murthy, Arthur T. Sands, and Michael T. Lotze

Nurix Therapeutics, San Francisco, CA, USA

254

Background

- *Ex-vivo* expanded autologous tumor-infiltrating lymphocyte (TIL) therapy is an adoptive cell therapy (ACT) that has demonstrated encouraging clinical responses in patients with melanoma and selected epithelial tumors.[1-6]
- Effective methods to obtain sufficient TIL with suitable quality and diversity from tumor samples remain a challenge, given the suppressive tumor microenvironment.[7]
- The E3 ubiquitin ligase, Casitas B lineage lymphoma B (CBL-B) is expressed in T cells where it regulates signaling through the T-cell receptor (TCR), limiting T-cell activation, differentiation, production of effector cytokines, and proliferation.
- Previous studies demonstrated that addition of NX-0255, a highly potent CBL-B inhibitor, during *ex-vivo* TIL expansion resulted in a favorable TIL phenotype and higher cell yields (**Figures 1 and 2**).[8]

Figure 1. CBL-B: A modulator of T-cell activation and novel target for immuno-oncology

- CBL-B E3 ligase is a master orchestrator of the immune response:
 - CBL-B knockout mice show robust T and NK cell-mediated anti-tumor activity

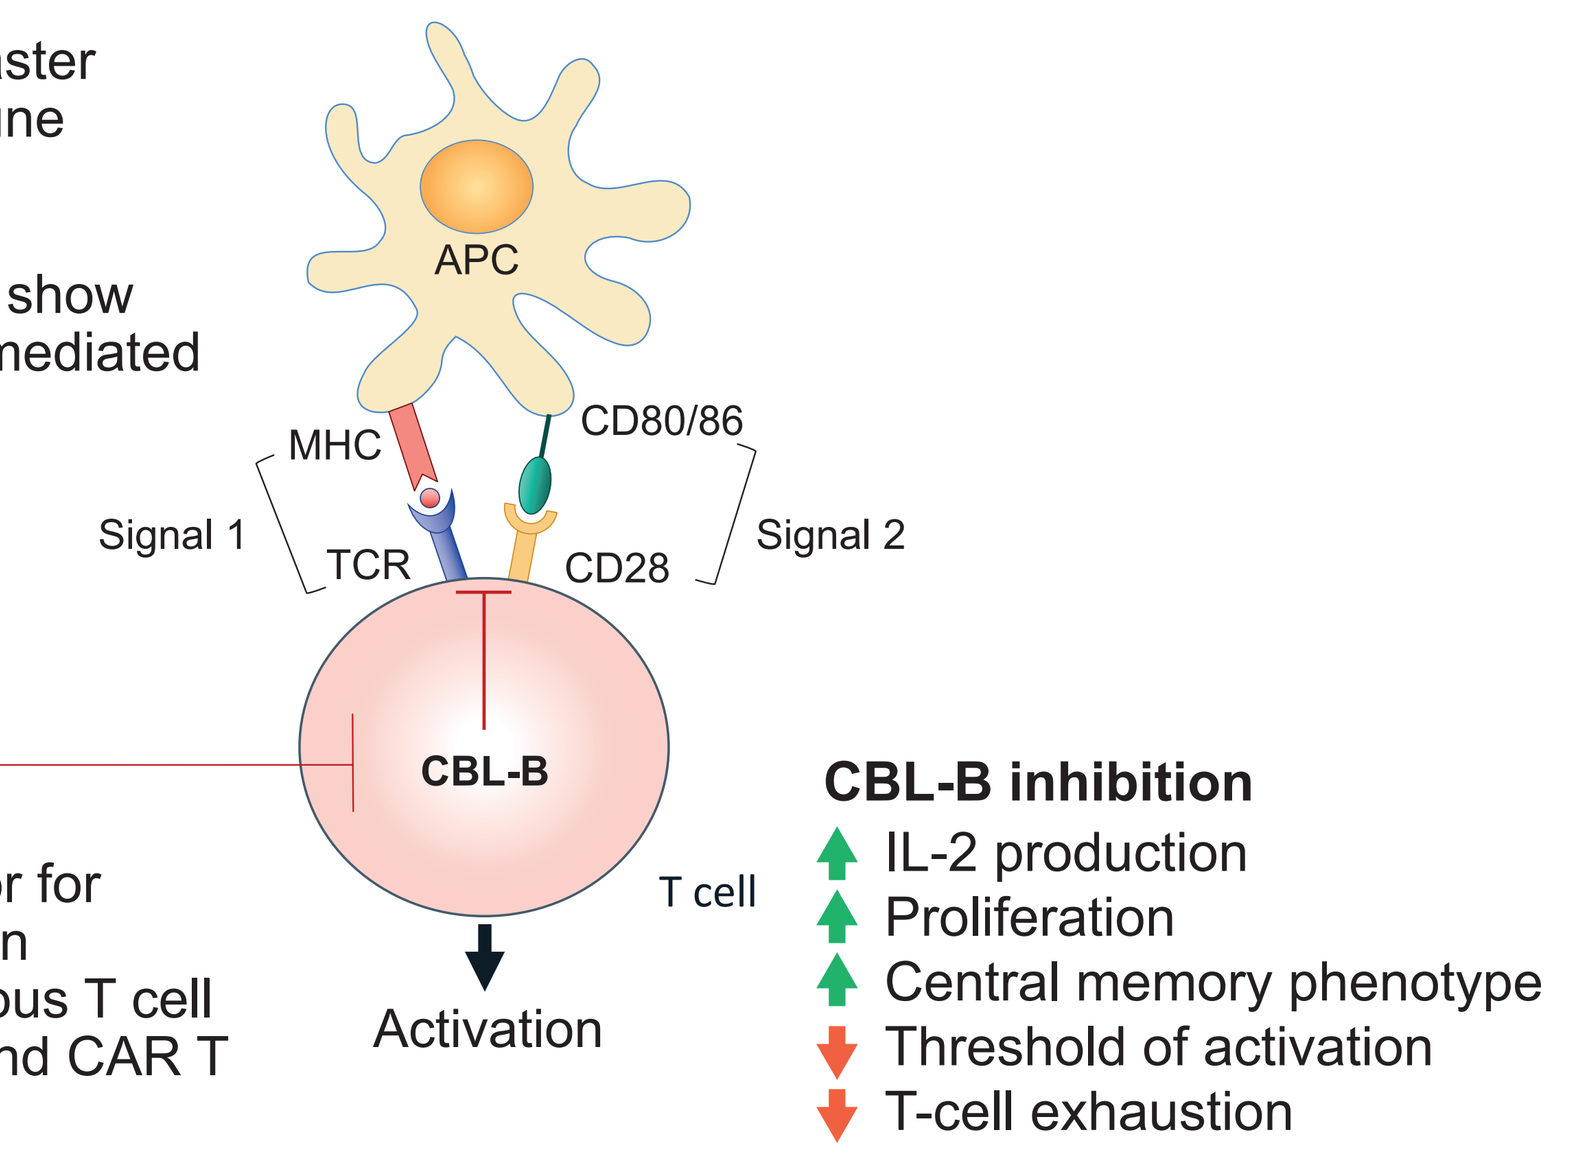
APC
MHC
CD80/86
Signal 1
Signal 2
TCR
CD28
CBL-B
T cell
NX-0255
Activation
CBL-B inhibition
IL-2 production
Proliferation
Central memory phenotype
Threshold of activation
T-cell exhaustion

Optimized CBL-B inhibitor for *ex vivo* use. Developing in conjunction with autologous T cell therapies including TIL and CAR T

Abbreviations: CBL-B, Casitas B-lineage lymphoma B; IL-2, interleukin-2; TCR, T-cell receptor; TIL, tumor-infiltrating lymphocytes

Figure 2. Overview of drug-enhanced TIL therapy

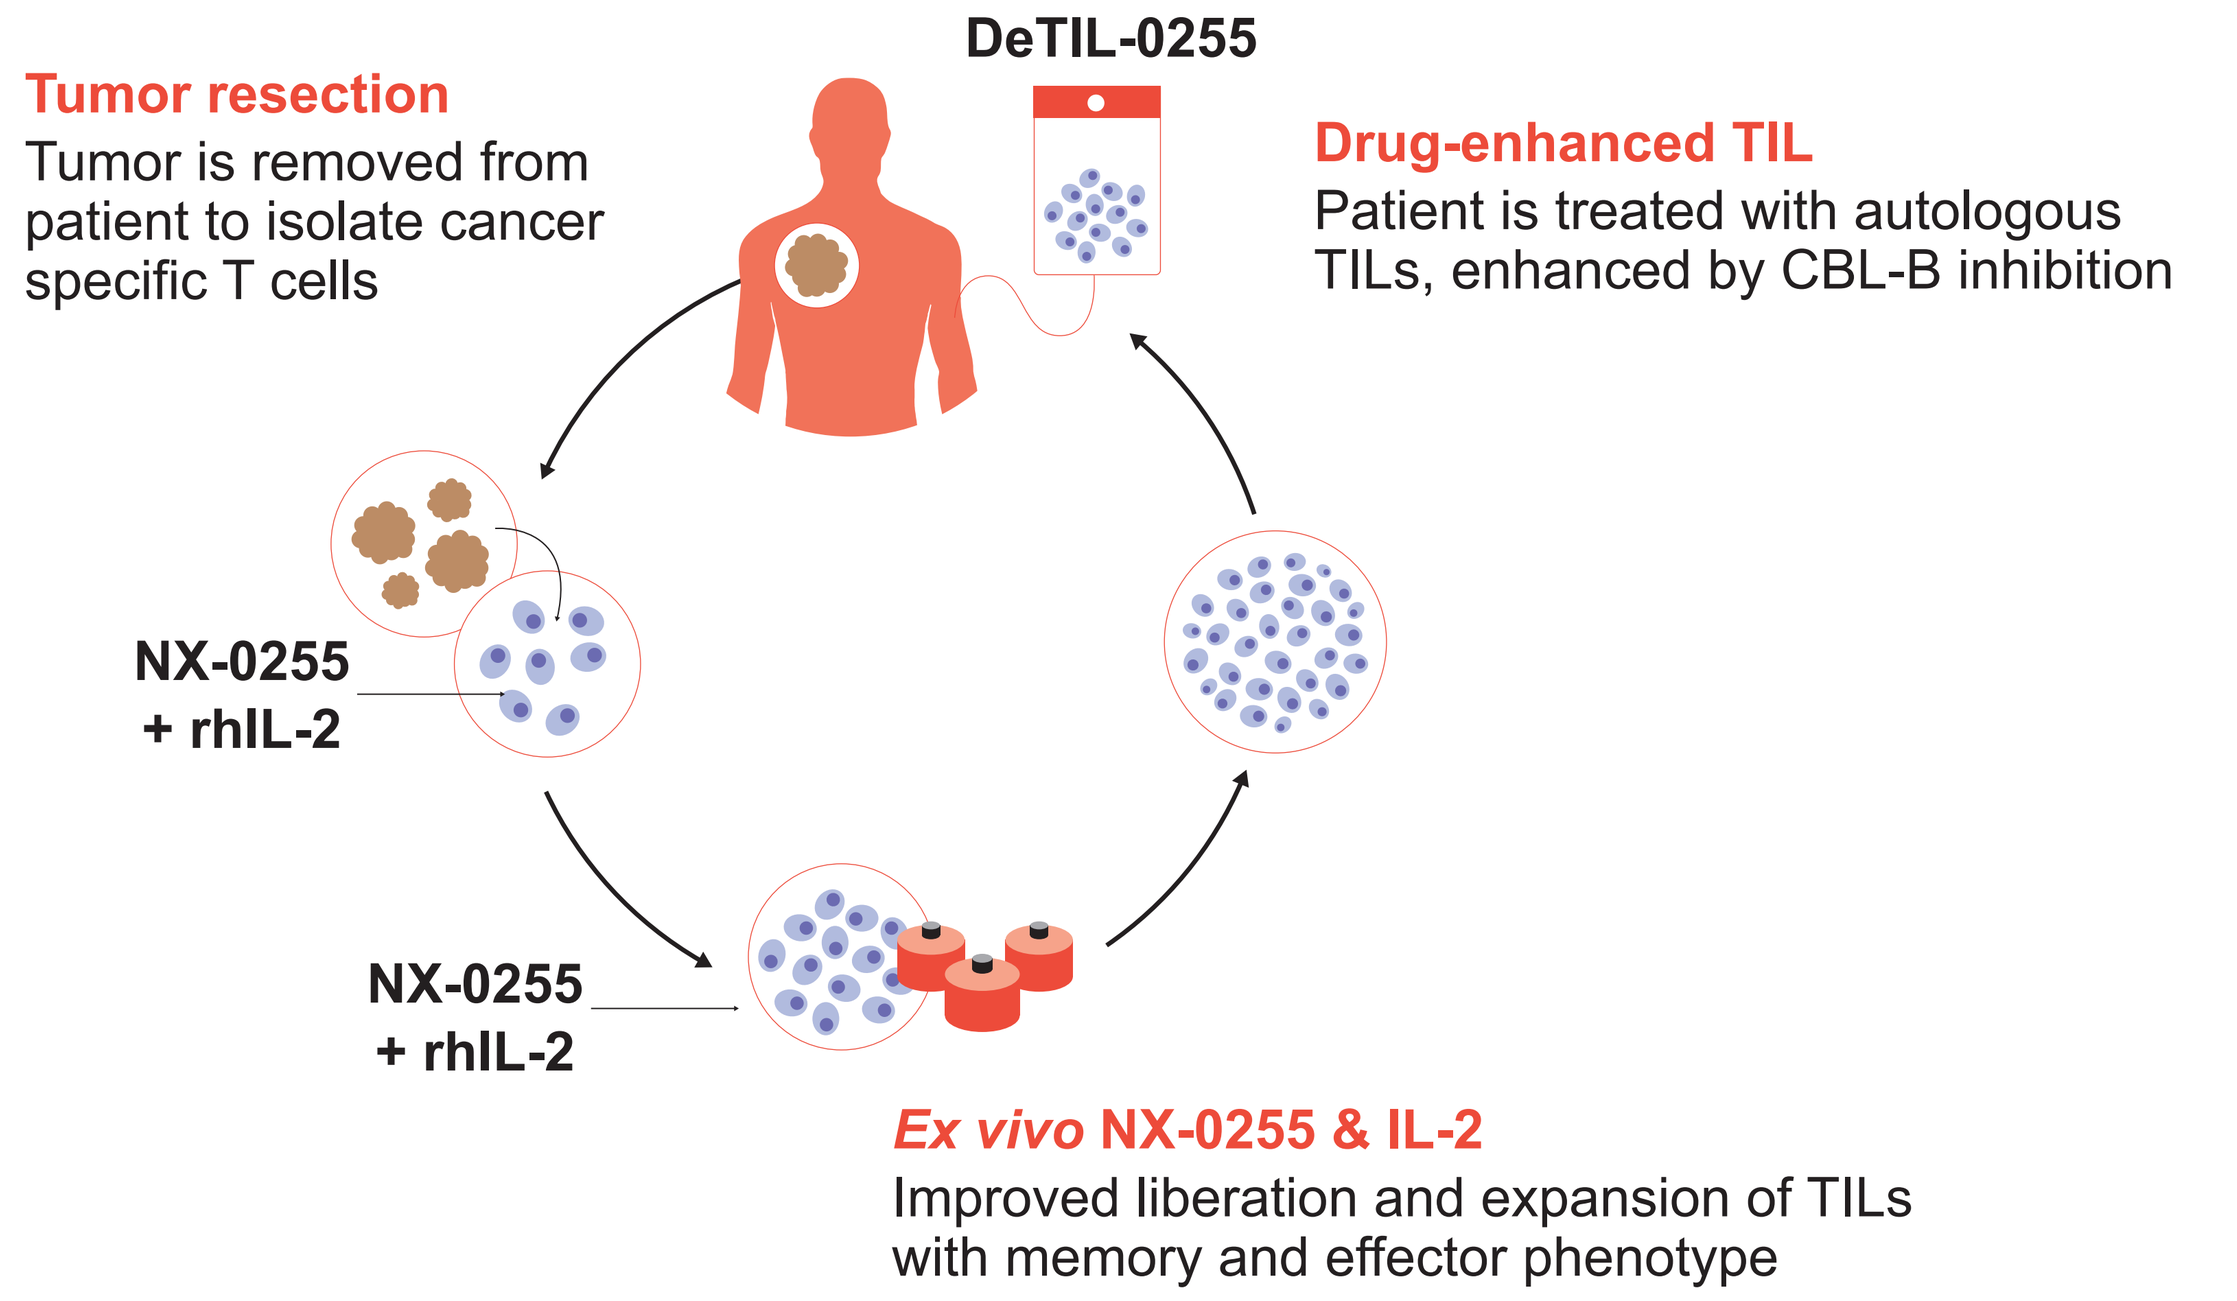
Tumor resection
Tumor is removed from patient to isolate cancer specific T cells
DeTIL-0255
Drug-enhanced TIL
Patient is treated with autologous TILs, enhanced by CBL-B inhibition
NX-0255 + rhIL-2
NX-0255 + rhIL-2
Ex vivo NX-0255 & IL-2
Improved liberation and expansion of TILs with memory and effector phenotype

Abbreviations: CBL-B, Casitas B lineage lymphoma-B; (rh)IL-2, (recombinant human) interleukin-2; TIL, tumor-infiltrating lymphocytes.

Objective

- To evaluate the effects of the CBL-B inhibitor NX-0255 on the expansion and phenotype of drug-enhanced TIL (DeTIL), in multiple full-scale (clinical scale manufacturing) processes.

Material and Methods

- Six full-scale studies were performed in parallel with TIL expanded:
 - in the presence of interleukin (IL)-2 (TIL arm).
 - in the presence of IL-2 and NX-0255 (DeTIL-0255 arm).
- Various tumor types were assessed, including endometrial (n=2), cervical (n=1), lung (n=1), colon (n=1), and melanoma (n=1) tumors.
- TIL and DeTIL-0255 were harvested following 22 days of expansion and assessed for total cell number, viability, phenotype, bulk transcriptome and function.

Statistical Analysis

- All statistical analyses were performed using a two-tailed Wilcoxon matched-pairs signed-rank test.

Results

Compared with the TIL arm, the addition of NX-0255 in DeTIL-0255:

- Demonstrated a trend towards an increased number of total viable cells (**Figure 3**).
- Increased the total number of $CD8^+$ T cells (**Figure 4**).
- Increased the total number of $CD8^+$ & $CD4^+$ central memory T cells (**Figure 5**).
- Increased the number of $CD8^+$ $CD39^-$ CD69- 'stem-like' memory cells (**Figure 6**).
- Increased the intracellular expression of interferon (IFN)-γ in response to phorbol-12-myristate-13-acetate (PMA) and Ionomycin stimulation and granzyme B, in response to αCD3-αCD28 stimulation (**Figure 7**).
- Enhanced granulysin, granzyme B and memory-related CD27 gene expression, suggesting an increased cytolytic potential and memory phenotype (**Figure 8**).

Figure 3. DeTIL-0255 demonstrate a trend towards an increased number of total viable cells

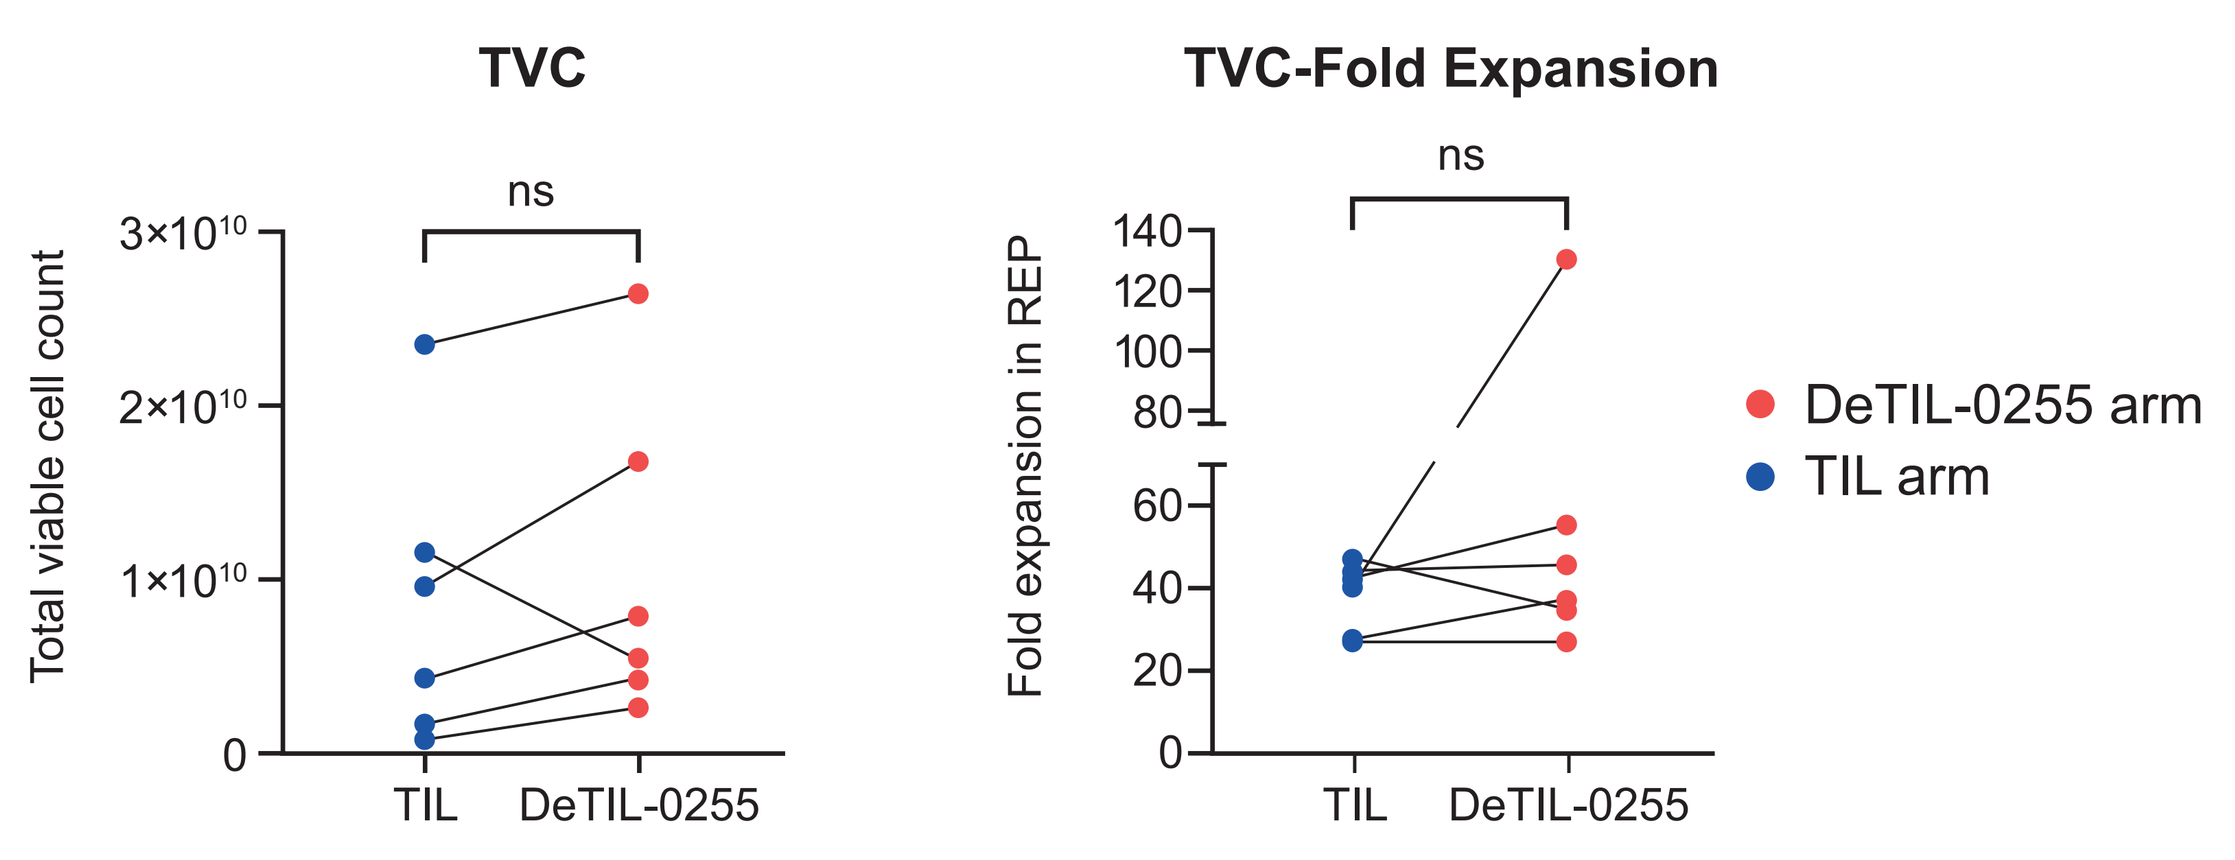
TVC
TVC-Fold Expansion
ns
ns
Total viable cell count
Fold expansion in REP
TIL
DeTIL-0255
DeTIL-0255 arm
TIL arm

ns, no significant difference
All cell counts were obtained following harvest and LOVO wash/concentration on Day 22 using the Chemometec NC-200. Fold expansion in the rapid expansion protocol was calculated as Day 22 total viable cells divided by Day 11 seeded cell numbers; DeTIL, drug-enhanced TIL; TIL, tumor-infiltrating lymphocyte; TVC, total viable cell.

Figure 4. The addition of NX-0255 enhances the total number of $CD8^+$ T cells

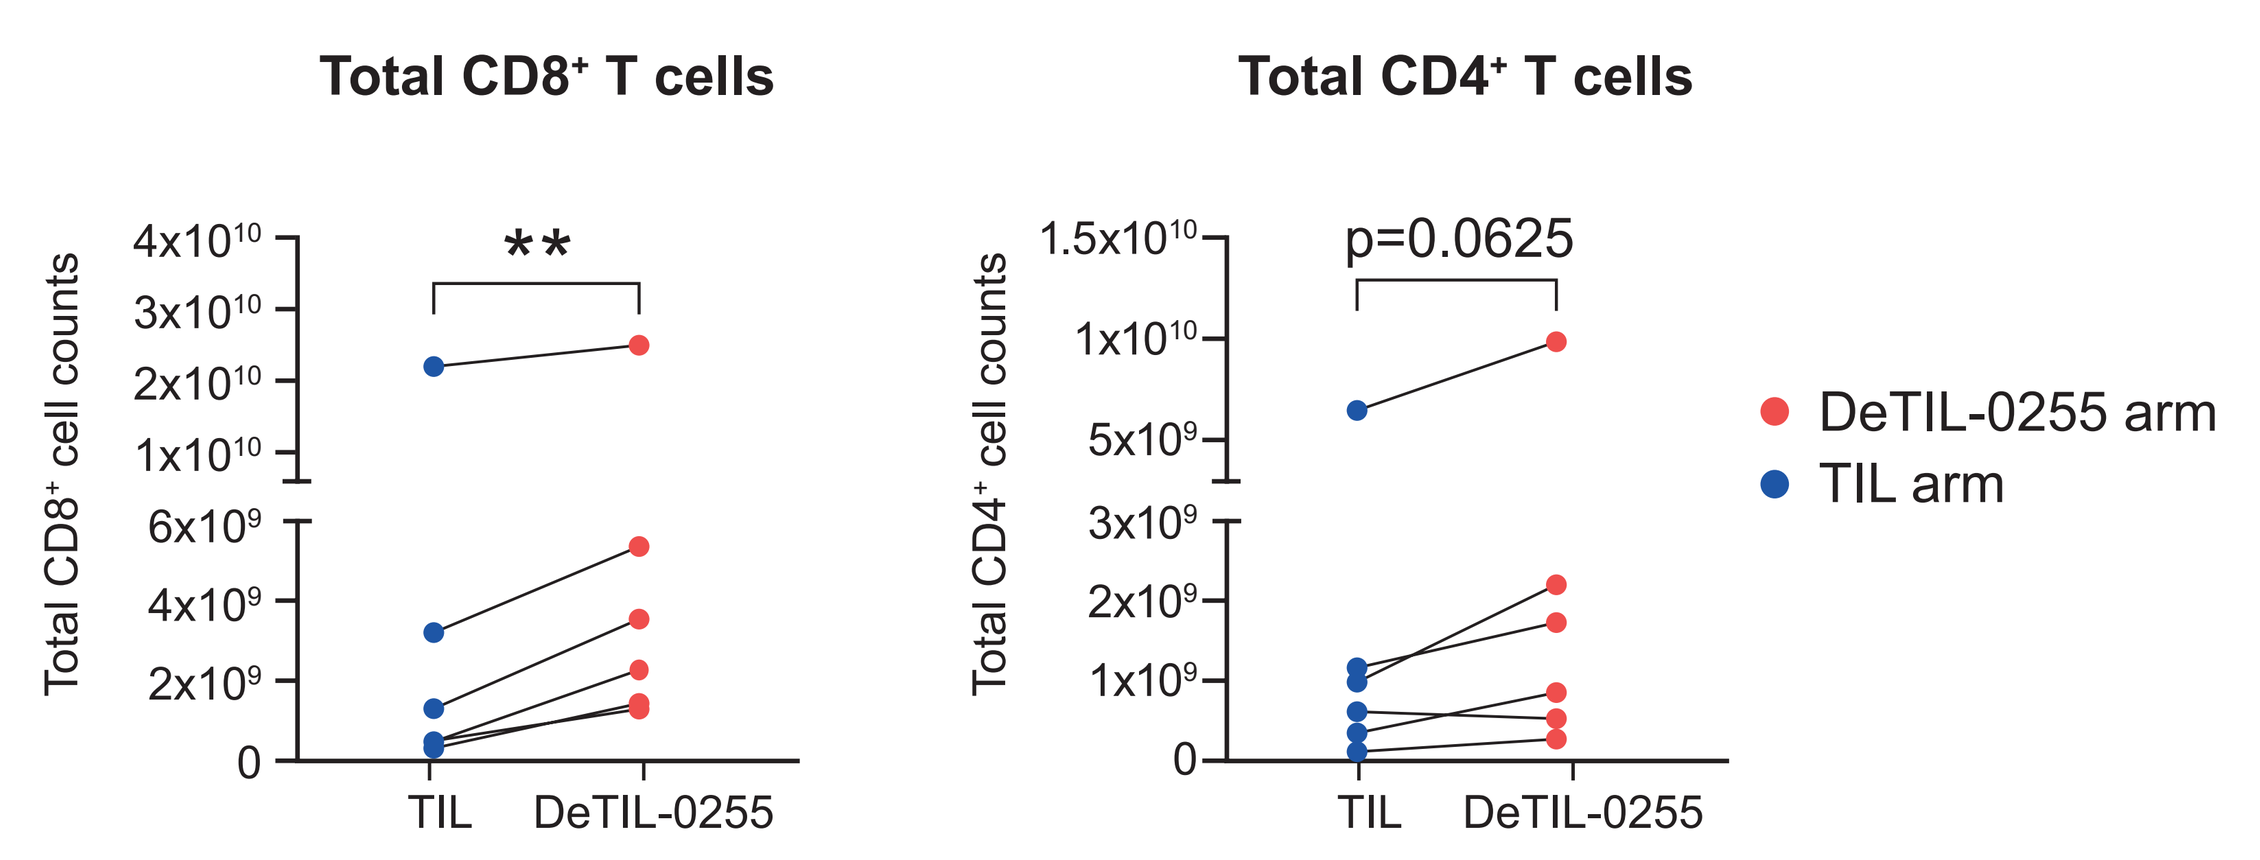
Total CD8+ T cells
Total CD4+ T cells
**
p=0.0625
Total CD8+ cell counts
Total CD4+ cell counts
TIL
DeTIL-0255
DeTIL-0255 arm
TIL arm

**p<0.01
The proportions of $CD8^+$ and $CD4^+$ T cells were assessed by flow cytometry. The absolute number of viable $CD8^+$ and $CD4^+$ T cells in TIL and DeTIL-0255 were determined for each individual culture. In all full-scale experiments, DeTIL-0255 demonstrated a significant increase in the total number of $CD8^+$ T cells and in five out of six experiments, an increase in the total numbers of $CD4^+$ T cells.
DeTIL, drug-enhanced TIL; TIL, tumor-infiltrating lymphocyte.

Figure 5. DeTIL-0255 demonstrate significantly increased $CD8^+$ and $CD4^+$ central memory T Cells

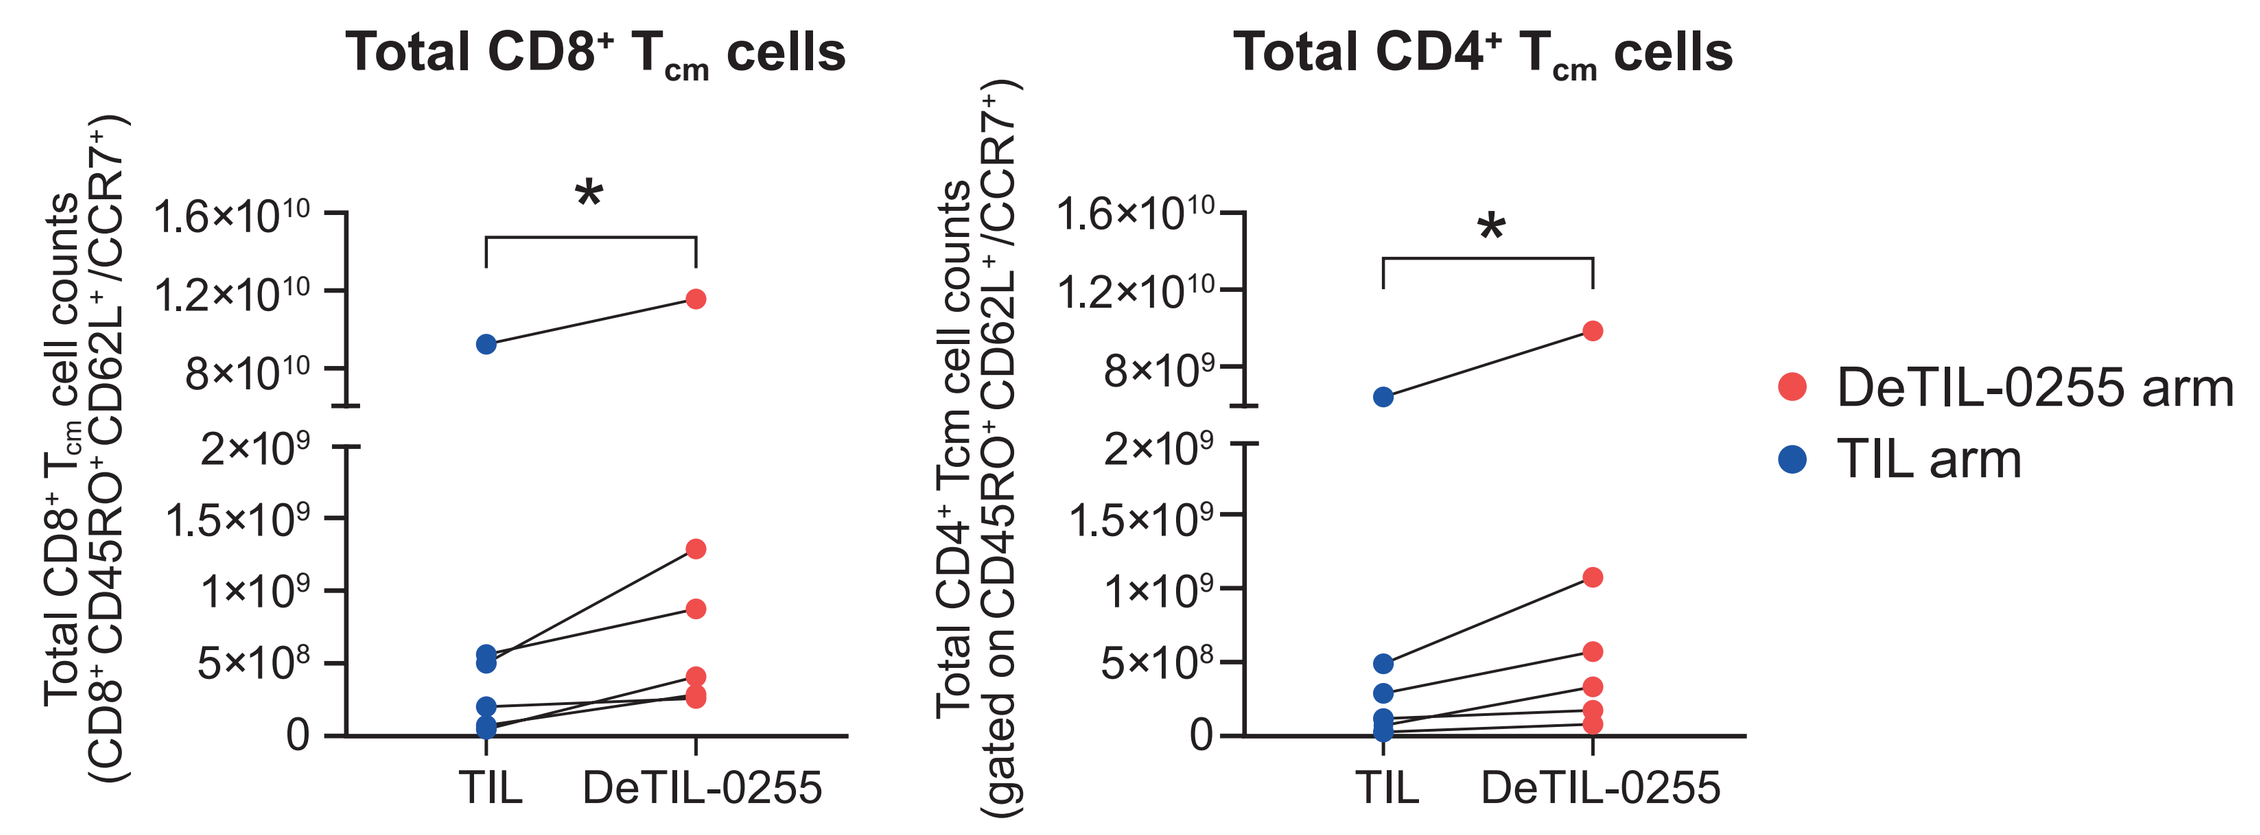
Total CD8+ Tcm cells
Total CD4+ Tcm cells
*
*
Total CD8+ Tcm cell counts (CD8+ CD45RO+ CD62L+/CCR7+)
Total CD4+ Tcm cell counts (gated on CD45RO+ CD62L+/CCR7+)
TIL
DeTIL-0255
DeTIL-0255 arm
TIL arm

*p<0.05
Central memory phenotype of $CD8^+$ or $CD4^+$ T cells was assessed by flow cytometry based on the percentages of $CD45RO^+$ $CD62L^+$ or $CD45RO^+$ $CCR7^+$. The absolute numbers of viable $CD8^+$ T_{CM} and $CD4^+$ T_{CM} cells were determined for each individual culture. All full-scale experiments showed significant increase in the total numbers of $CD8^+$ and $CD4^+$ T central memory T cells.
DeTIL, drug-enhanced TIL; T_{cm}, central memory T cell; TIL, tumor-infiltrating lymphocyte.

Figure 6. DeTIL-0255 demonstrate significantly increased $CD39^-$ $CD69^-$ stem-like $CD8^+$ T-cells when compared with TIL

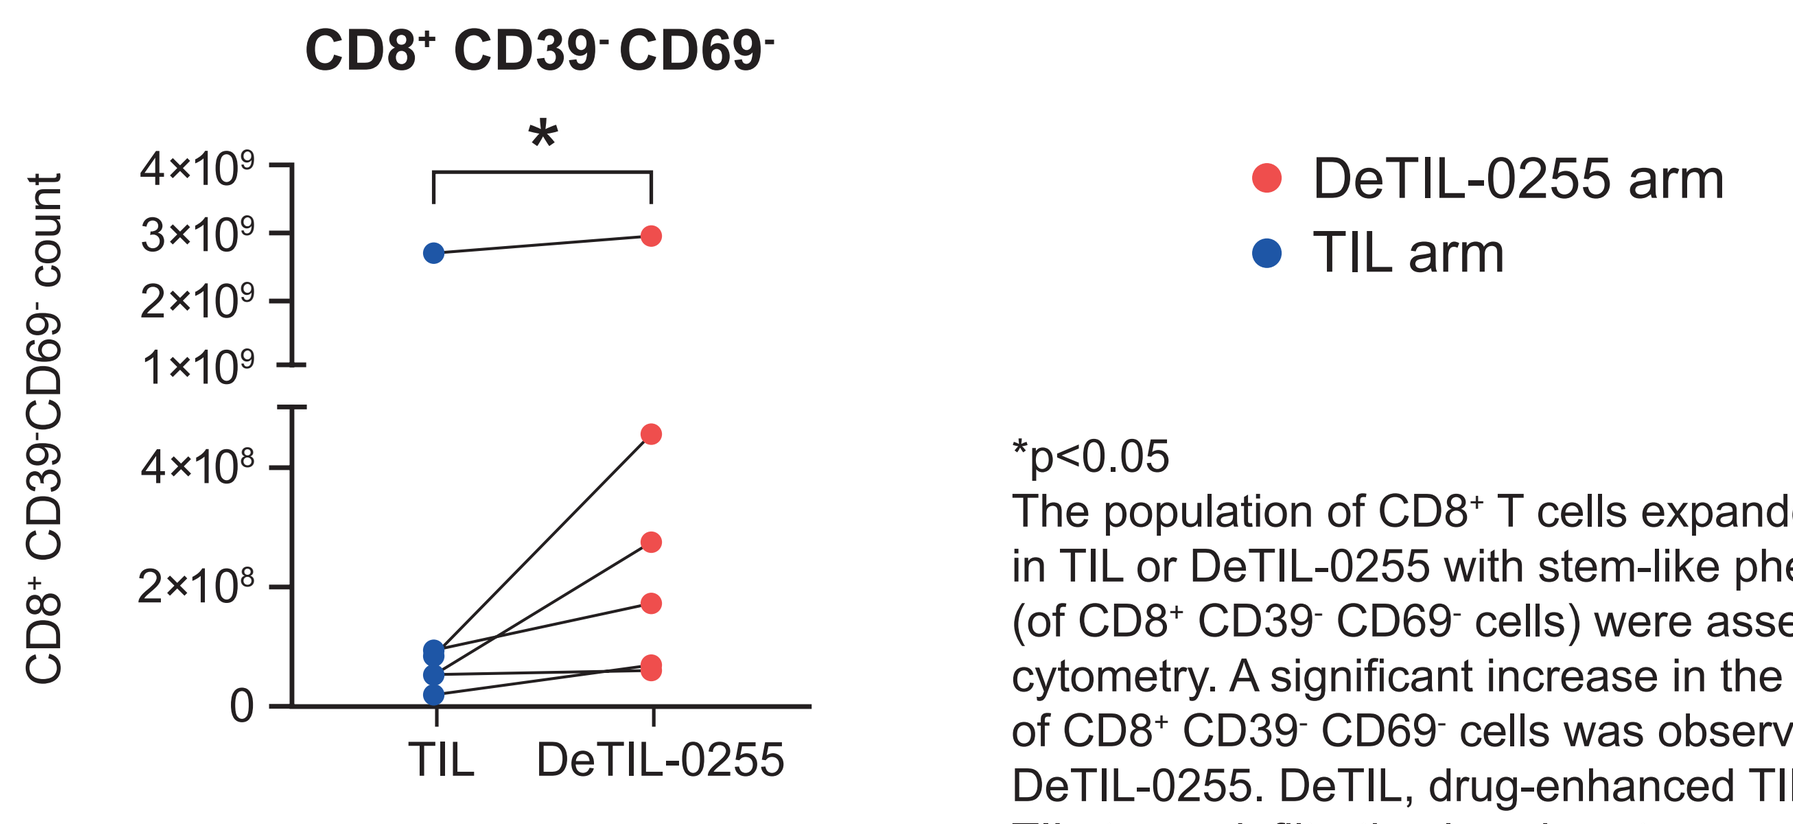
CD8+ CD39- CD69-
*
CD8+ CD39-CD69- count
TIL
DeTIL-0255
DeTIL-0255 arm
TIL arm

*p<0.05
The population of $CD8^+$ T cells expanded for 22 days in TIL or DeTIL-0255 with stem-like phenotype markers (of $CD8^+$ $CD39^-$ $CD69^-$ cells) were assessed by flow cytometry. A significant increase in the total number of $CD8^+$ $CD39^-$ $CD69^-$ cells was observed in the DeTIL-0255. DeTIL, drug-enhanced TIL; TIL, tumor-infiltrating lymphocyte.

Figure 7. DeTIL-0255 demonstrate significantly increased proportions of $CD8^+$ T cells expressing intracellular IFN-γ and granzyme B following stimulation

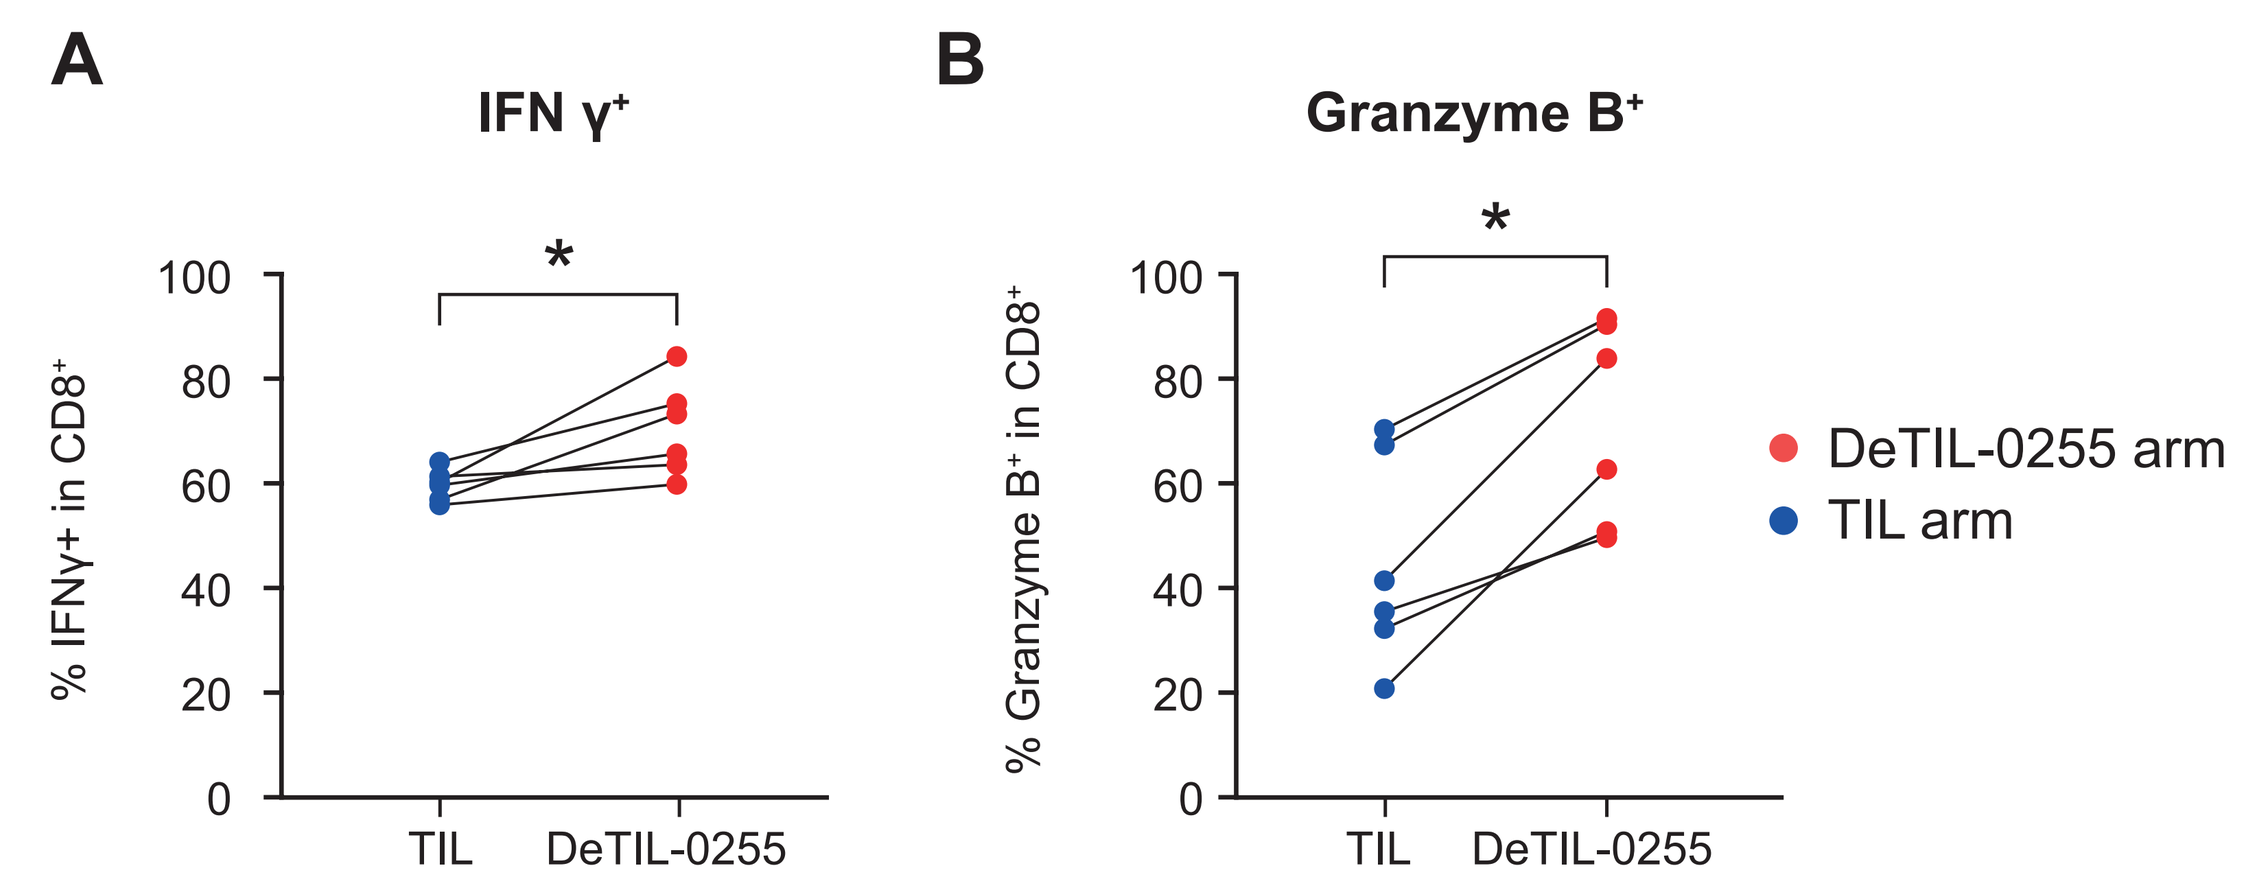
A
IFN γ+
B
Granzyme B+
*
*
% IFNγ+ in CD8+
% Granzyme B+ in CD8+
TIL
DeTIL-0255
DeTIL-0255 arm
TIL arm

*p<0.05
Expanded TIL and DeTIL-0255 were stimulated for 6 hours with (**A**) PMA and Ionomycin or (**B**) anti-CD3 and anti-CD28 in the presence of Monensin and Brefeldin A. After 6 hours, expression of intracellular cytotoxic markers, IFN-γ and granzyme B were assessed by flow cytometry. Significant differences in the percentage of $CD8^+$ T cells expressing IFN-γ (**A**) and Granzyme B (**B**) were observed following stimulation in DeTIL-0255 compared with TIL. DeTIL, Drug-enhanced TIL; IFN, interferon; TIL, tumor-infiltrating lymphocyte.

Figure 8. DeTIL-0255 demonstrate an increased cytotoxic and memory gene expression profile

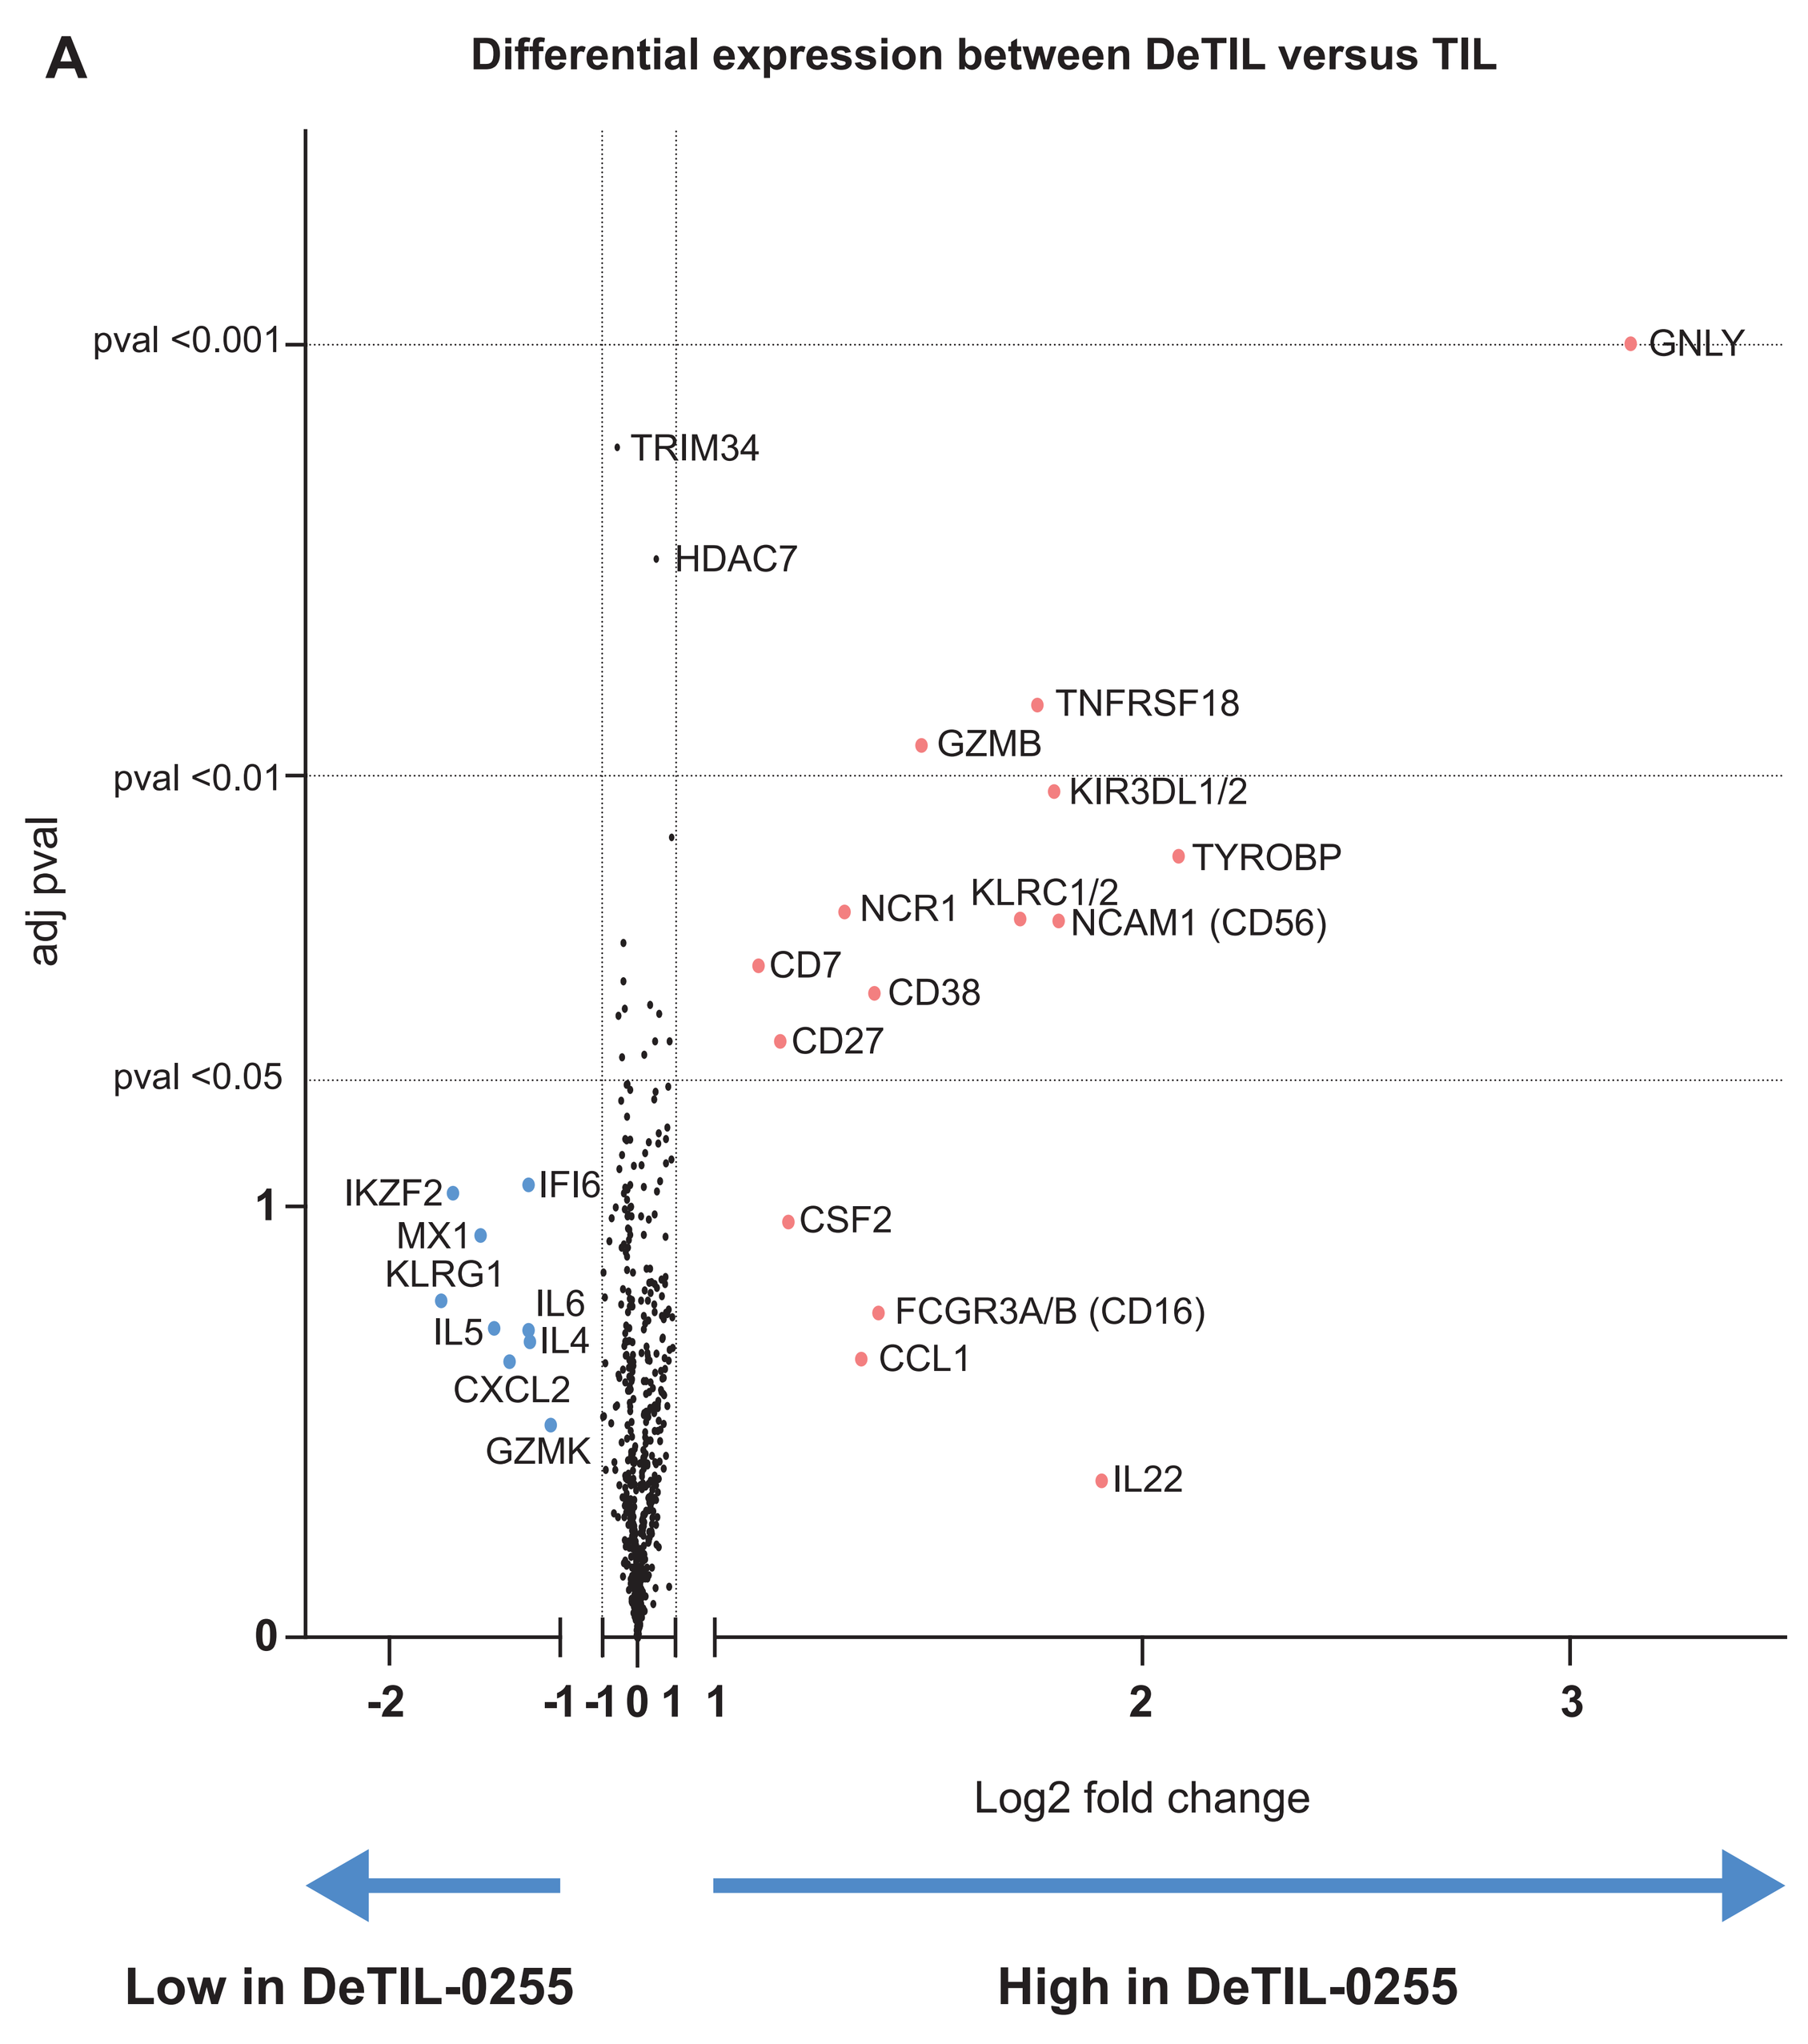
A
Differential expression between DeTIL versus TIL
pval <0.001
pval <0.01
pval <0.05
adj pval
GNLY
TRIM34
HDAC7
TNFRSF18
GZMB
KIR3DL1/2
TYROBP
NCR1
KLRC1/2
NCAM1 (CD56)
CD7
CD38
CD27
IKZF2
IFI6
MX1
KLRG1
IL6
IL5
IL4
CXCL2
GZMK
CSF2
FCGR3A/B (CD16)
CCL1
IL22
Log2 fold change
Low in DeTIL-0255
High in DeTIL-0255

Figure 8 (cont.) DeTIL-0255 demonstrate an increased cytotoxic and memory gene expression profile

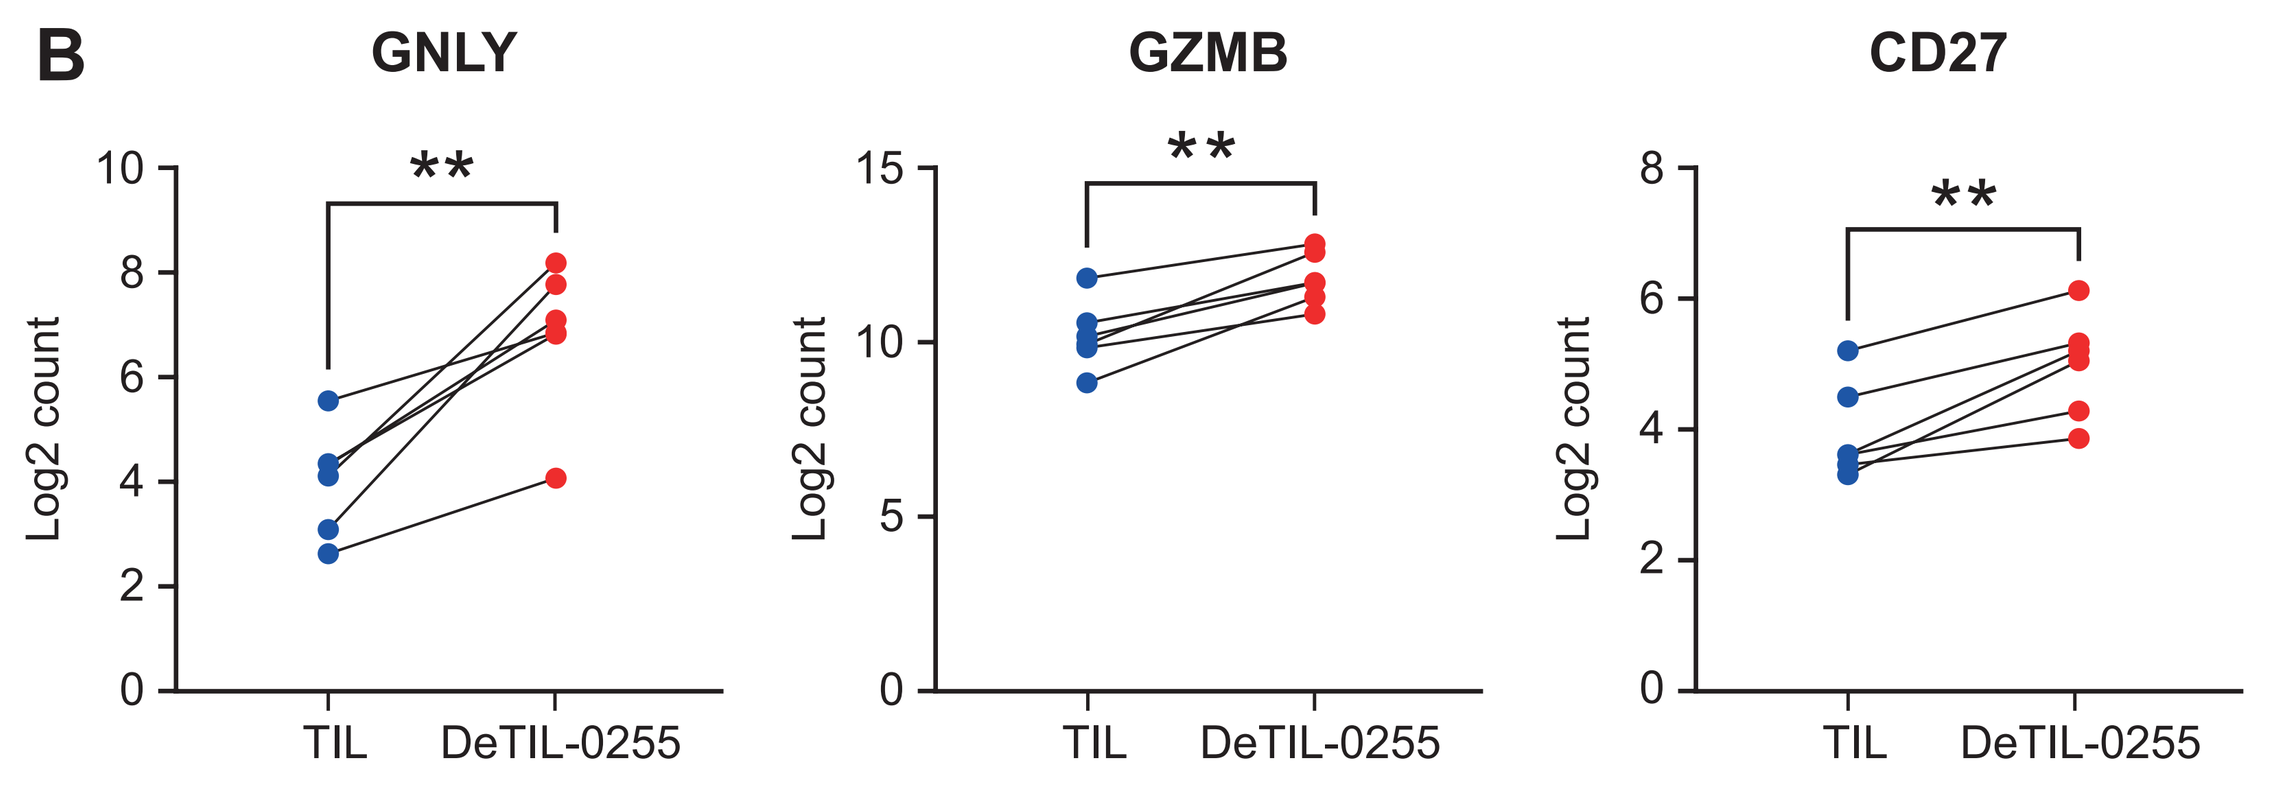
B
GNLY
GZMB
CD27
**
**
**
Log2 count
TIL
DeTIL-0255

* p<0.05, ** p<0.01
Gene expression profile of DeTIL-0255 and TIL were evaluated by Nanostring and analyzed by nSolver analysis software. (**A**) Differential expression between DeTIL-0255 and TIL. (**B**). Log2 probe counts confirm Increase in cytotoxic gene Granulysin and Granzyme B, and memory related gene CD27 expression.
DeTIL, Drug-enhanced TIL; GNLY, granulysin; GZMB, granzyme B; TIL, tumor-infiltrating lymphocyte.

Conclusion

- Manufacturing of DeTIL-0255 is feasible, and compared with conventional TIL, demonstrates a superior phenotype and function that is suitable for clinical evaluation.
- A clinical trial has been initiated with DeTIL-0255 in patients with gynecologic malignancies (ovarian cancer, endometrial cancer, and cervical cancer; NCT05107739).

References

1. Rosenberg SA, et al. Clin Cancer Res. 2011;17:4550–7.
2. Zacharakis N, et al. Nat Med 2018; 24, 724–30.
3. Wang S, et al. BMC Medicine 2021,19:140–7.
4. Krishna S, et al. Science 2020;370:1328–34.
5. Lutz-Micoladoni C, et al. Frontiers in Oncology 2015;5:1–14.
6. Zhou X, et al. Signal Transduction And Targeted Therapy 2021;6:16.
7. Soloff AC, Lotze MT. Proc Natl Acad Sci U S A. 2019;116,22901–3.
8. Whelan S, et al. Poster presented at the 36th SITC Annual Meeting, Washington DC, USA; November 12–14, 2021.

Acknowledgements

Research was sponsored by Nurix Therapeutics, Inc. Medical writing was provided by Xiaoyan Liang of Nurix Therapeutics, Inc.. Editorial support was provided by Miller Medical Communications, which was funded by Nurix Therapeutics, Inc.

Other DeTIL-0255 posters presented at SITC 2022

#671 Girda E, et al. A phase 1 adoptive cell therapy using drug-enhanced, tumor-infiltrating lymphocytes, DeTIL-0255, in adults with advanced malignancies

#361 Murthy P, et al. Universal expansion of CBL-B–inhibited tumor-infiltrating lymphocytes, DeTIL-0255, from women with ovarian cancer: process validation

#331 Gallotta M, et al. A novel small molecule inhibition of CBL-B shows potent antitumor activity in combination with Pmel-1 adoptive cell transfer in an aggressive mouse melanoma model

Presented at the SITC 37th Annual Meeting, November 10–12, 2022, Boston, MA, USA