Ex-vivo inhibition of CBL-B with a novel small molecule inhibitor, NX-0255, enhances persistence and anti-tumor activity of adoptively transferred CD8+ T cells in mouse tumor models

Abstract Number 573

Marilena Gallotta*, Jose Gomez Romo*, Sasha Borodovsky*, Austin Tenn-McClellan*, Jennifer Stokes*, Serena Ranucci*, Jennifa Gosling*, Gwenn M Hansen*, Arthur Sands*, Ryan Rountree*, Cristiana Guiducci*

**Nurix Therapeutics, San Francisco, CA, USA*

Abstract

Adoptive cell transfer (ACT) involving engineered T cells or autologous tumor-specific lymphocytes (TIL) has shown promising results in the treatment of advanced cancer. Poor in vitro cell expansion, inefficient T-cell migration and T cell exhaustion in the tumor microenvironment greatly limits the broader application of this approach. The E3 ubiquitin ligase Casitas B-lineage lymphoma b (CBL-B) is highly expressed in T cells, where it constrains cell activation following TCR engagement, therefore functioning as an important intracellular checkpoint that limits T cell mediated anti-tumor responses. We have developed NX-0255, a highly potent small molecule inhibitor of CBL-B, that increases TCR-mediated activation of T cells in the presence or absence of CD28 co-stimulation.

Here we used the OT-1 ACT/E.G7-OVA lymphoma and the Pmel-1 ACT/B16 melanoma tumor models to investigate whether adding NX-0255 during the *in vitro* expansion of tumor-specific T cells would have favorable effects on their ability to reject tumors following adoptive transfer *in vivo*. OT-1 CD8+ T cells were treated *in vitro* using anti-CD3 stimulation combined with either NX-0255 alone, IL-2 alone, or NX-0255 and IL-2, and subsequently transferred *in vivo* to mice bearing established E.G7-OVA. ACT of OT-1 CD8+ T cells cultured with both NX-0255 plus IL-2 demonstrated superior anti-tumor activity against E.G7-OVA lymphoma tumors, resulting in significant improvement in survival when compared to mice receiving OT-1 CD8+ T cells cultured with NX-0255 alone or IL-2 alone. Importantly, OT-1 CD8+ T cells cultured in the presence of NX-0255 or the combination of NX-0255 plus IL-2 were found at an increased frequency in the blood and in the tumor and persisted longer in circulation when compared to cells cultured with IL-2 alone. A lower percentage of the tumor-infiltrating OT-1 cells were triple-positive for exhaustion markers PD1+TIM3+LAG3+ expression when cultured with NX-0255 alone or the combination of NX-0255 and IL-2 compared to culture with IL-2 alone.

Similarly, ACT of Pmel-1 CD8+ T cells cultured with the combination of NX-0255 plus IL-2 provides a robust and durable anti-tumor response compared to mice receiving Pmel-1 CD8+ T cells cultured with IL-2 alone in the aggressive B16-OVA model. Flow cytometry analysis performed at 13 and 26 days after ACT showed that Pmel-1 CD8+ T cells cultured in the presence of NX-0255 plus IL-2 were found at an increased frequency in the blood and showed an increased central-memory phenotype (CD44+CD62L+) after adoptive transfer when compared to cells cultured with IL-2 alone. Collectively, these data suggest that adding the CBL-B inhibitor, NX-0255, during the *in vitro* expansion of tumor-specific T cells increases the frequency and absolute numbers of less exhausted CD8+ memory T-cells, profoundly affecting their functionality, increasing their in vivo persistence and ability to infiltrate the tumor.

The resulting enhancement of anti-tumor activity supports the rationale for the use of NX-0255 in the production of an investigational drug-enhanced TIL therapy, DeTIL-0255, which is currently in a Phase 1 clinical trial [NCT05107739].

Results

Figure 1. NX-0255 treated OT-I cells differentiate into potent effectors capable of rejecting established tumors

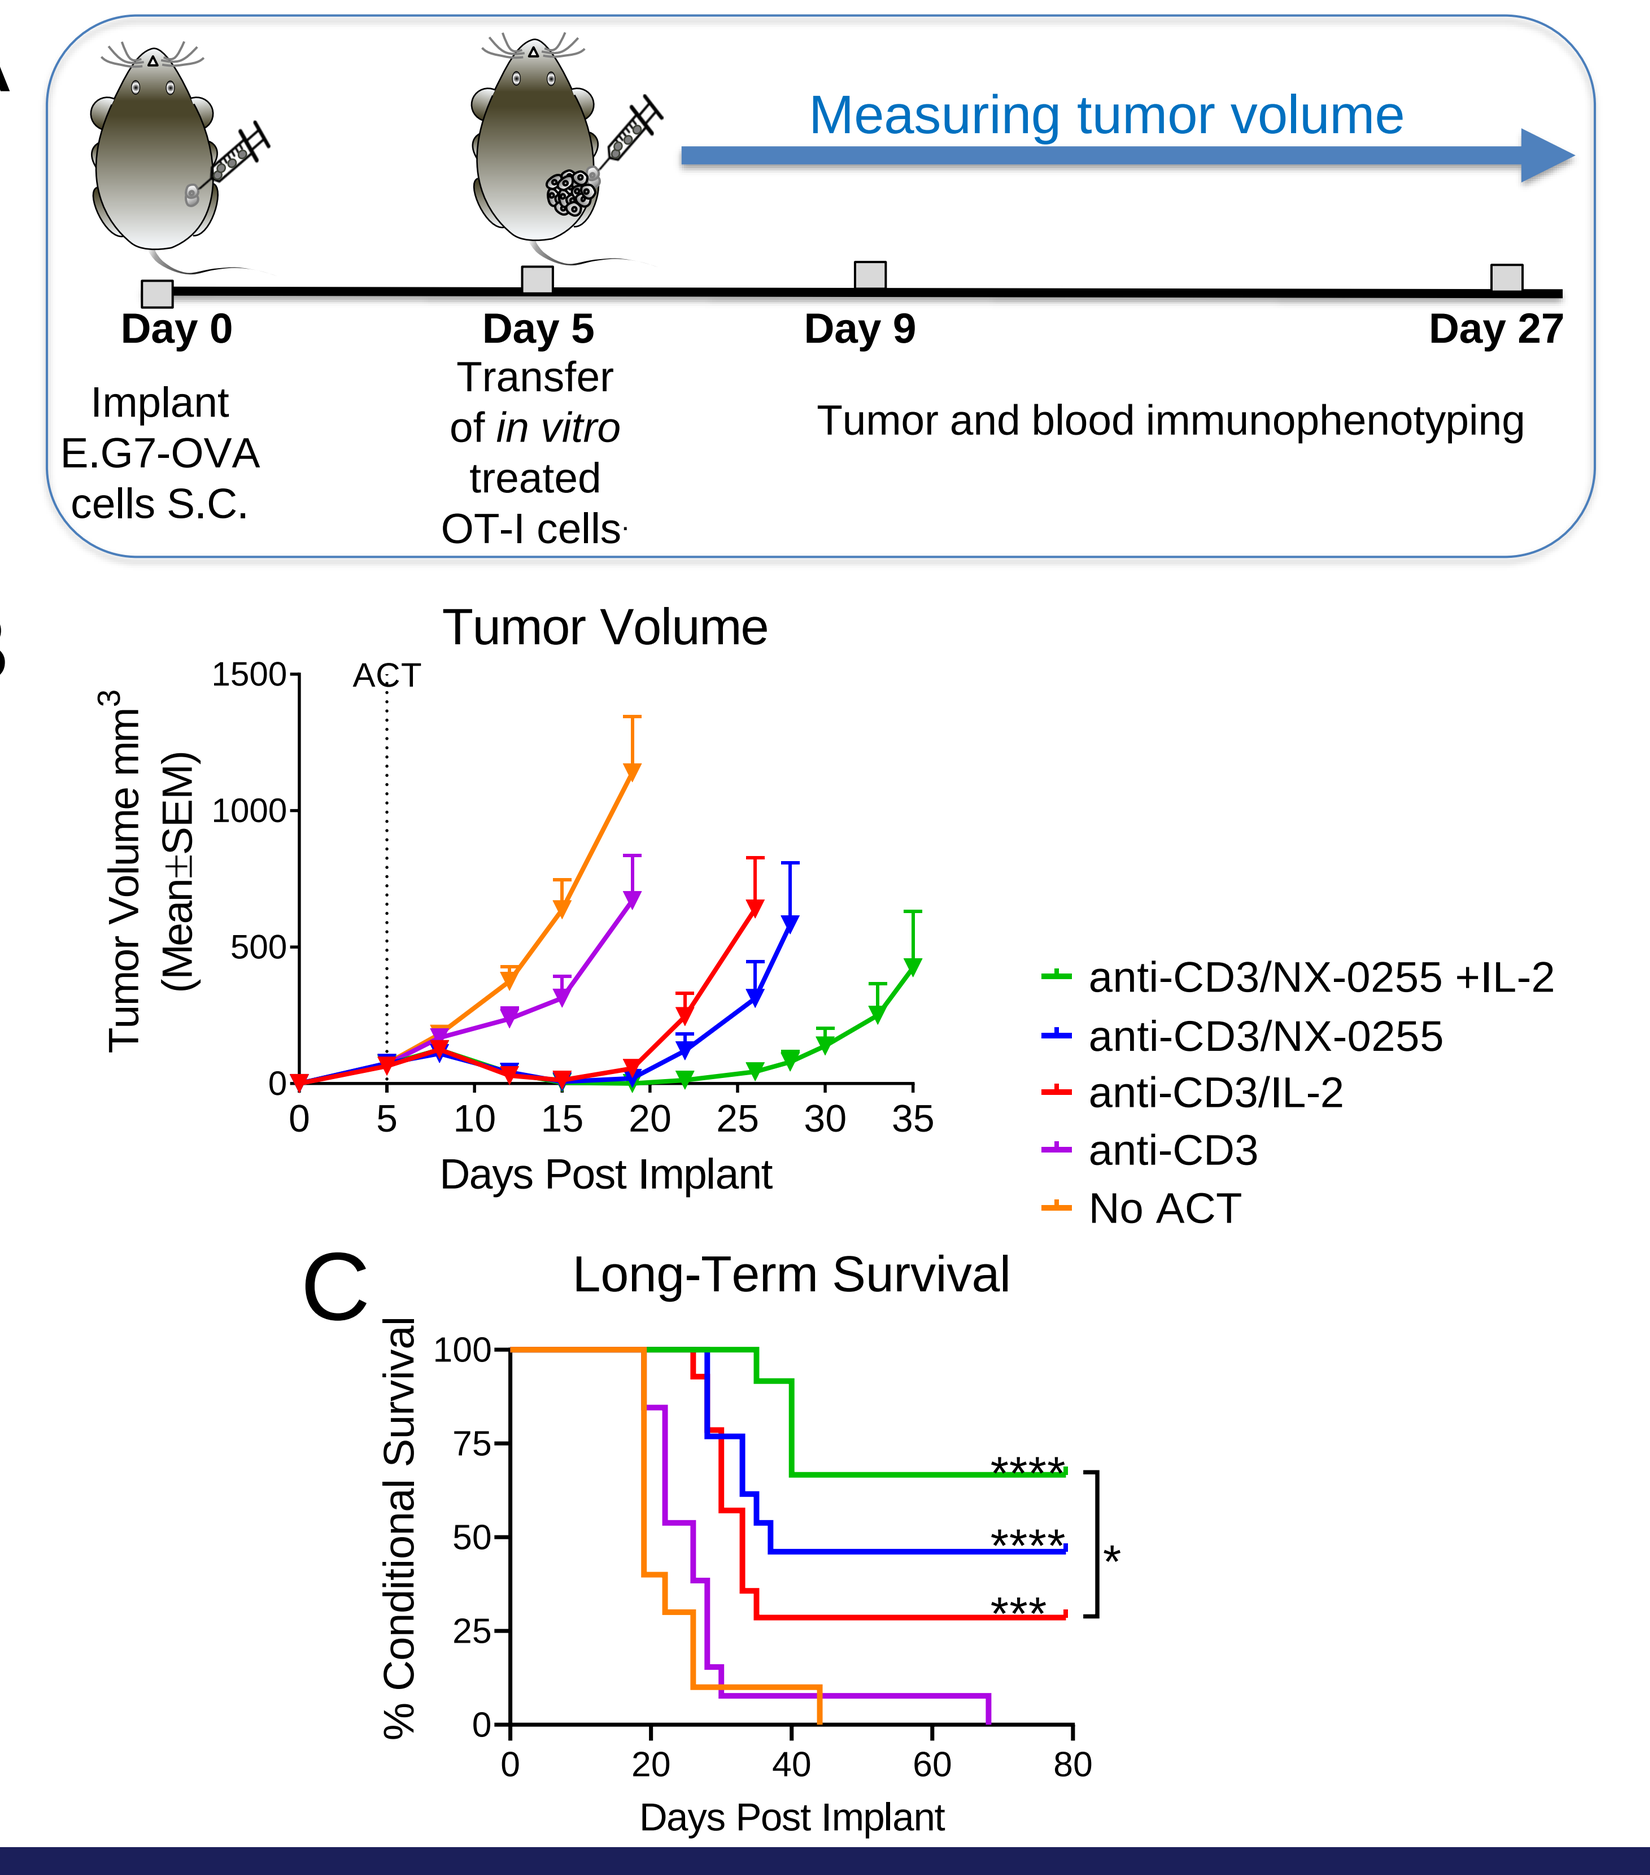
A
Measuring tumor volume
Day 0
Day 5
Day 9
Day 27
Implant E.G7-OVA cells S.C.
Transfer of *in vitro* treated OT-I cells
Tumor and blood immunophenotyping
B
Tumor Volume
ACT
Tumor Volume mm3 (Mean±SEM)
Days Post Implant
anti-CD3/NX-0255 +IL-2
anti-CD3/NX-0255
anti-CD3/IL-2
anti-CD3
No ACT
C
Long-Term Survival
% Conditional Survival
Days Post Implant

(A-C) Mice bearing E.G7-OVA s.c. tumors were treated IV on Day 5 with ACT of CD3-stimulated OT-I CD8+ T cells that were treated in the presence of either: 300 IU/mL IL-2; 1 µM NX-0255; 1 µM NX-0255 and 300 IU/mL IL-2; or no additional agents (CD3 stimulation alone). No ACT control mice were administered only media IV. Figure 1A shows a schematic of the study and sampling time points. Figure 1B shows mean tumor volumes ± SEM, on Days 0 through 35; lines end for each group at the last day that all animals designated for tumor volume and long-term survival assessment were alive. Dashed line marks Day 5 when ACT was administered. Figure 1C shows the percentage of animals surviving between Days 0 to 79 as defined by reaching the conditional survival endpoint of either bearing a tumor with volume exceeding size limits set or having met a humane endpoint. Statistical significance of differences in survival between groups was evaluated using the Log-rank (Mantel-Cox) test (* P ≤ 0.05, *** P ≤ 0.001, and **** P ≤ 0.0001).

Results (Continued)

Figure 2. NX-0255 treated OT-I cells show increased expansion in tumor and blood and decreased surface expression of exhaustion markers

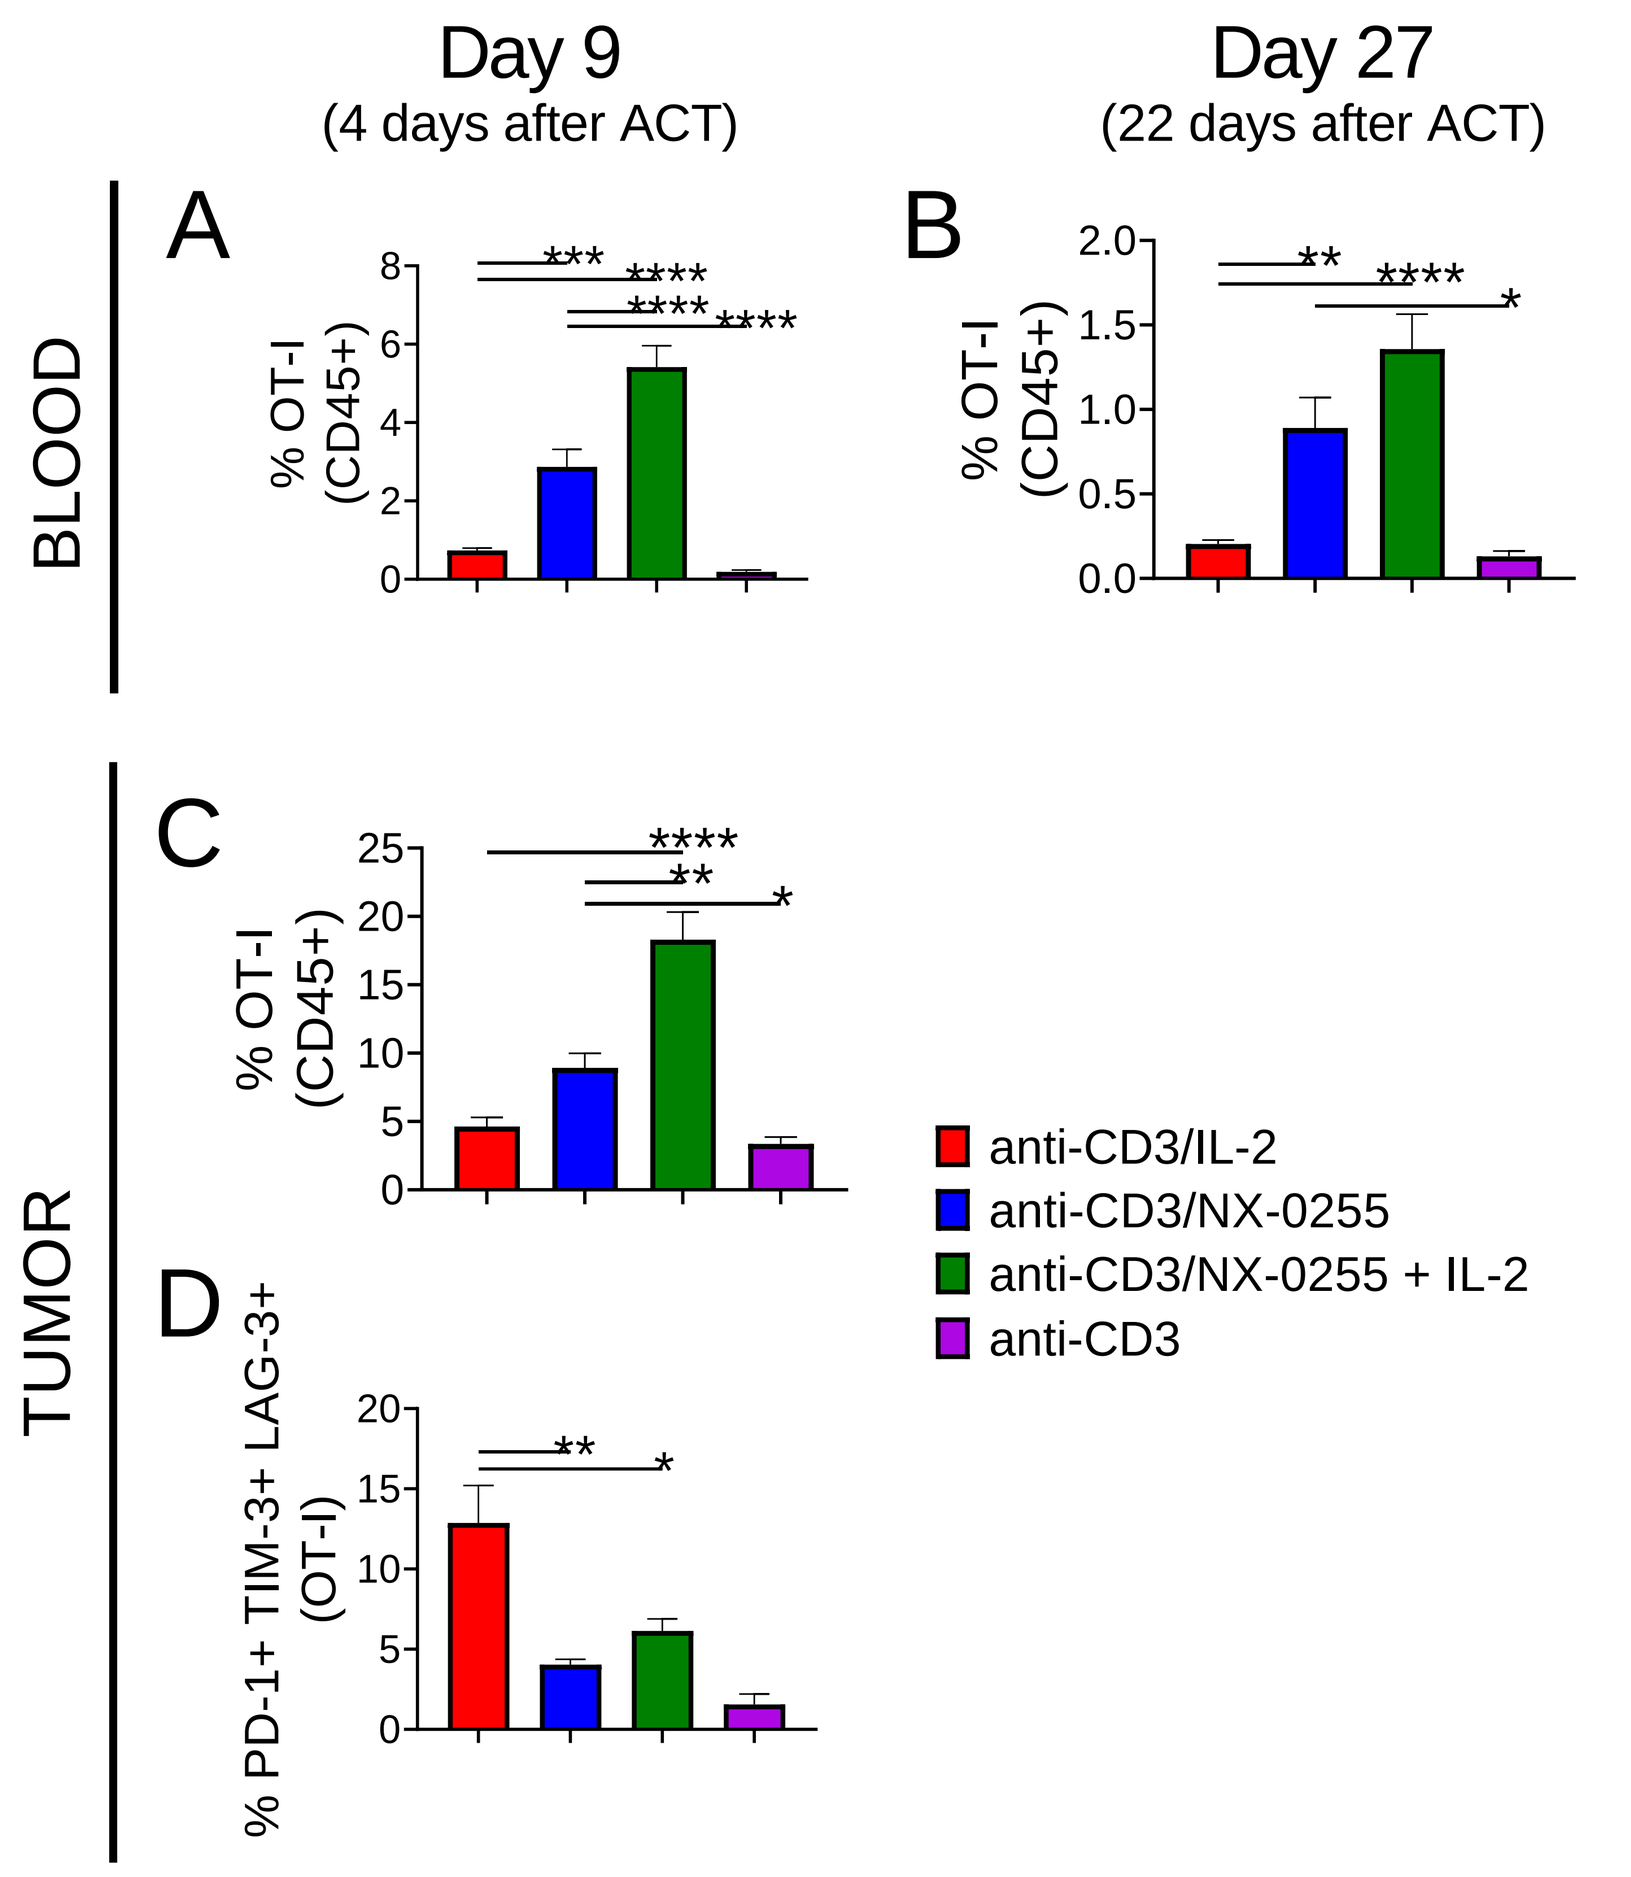
Day 9
(4 days after ACT)
Day 27
(22 days after ACT)
BLOOD
TUMOR
A
B
C
D
% OT-I (CD45+)
% PD-1+ TIM-3+ LAG-3+ (OT-I)
anti-CD3/IL-2
anti-CD3/NX-0255
anti-CD3/NX-0255 + IL-2
anti-CD3

(A-D) Mice bearing E.G7-OVA s.c. tumors were treated IV on Day 5 with ACT of CD3-stimulated OT-I CD8+ T cells that were treated in the presence of either: 300 IU/mL IL-2; 1 µM NX-0255; 1 µM NX-0255 and 300 IU/mL IL-2; or no additional agents (CD3 stimulation alone). No ACT control mice were administered only media IV. On Day 9, corresponding to 4 days after ACT, and Day 27, corresponding to 22 days after ACT, blood was collected from all available animals and the frequency of OT-I CD8+ T cells was determined as a percentage of CD45+ leukocytes by flow cytometry (A, Day 9; B, Day 27). Also on Day 9, tumors were collected from five mice euthanized in each of the groups and the frequency of OT-I CD8+ T cells was determined as a percentage of CD45+ leukocytes (C) or the percentage of triple-positive OT-I CD8+ T cells was determined for the exhaustion markers PD-1, TIM-3, and LAG-3 (D). Group mean values ± SEM are shown in each graph. Statistical significance was determined by one-way ANOVA with Tukey's multiple comparisons test (* P ≤ 0.05, ** P ≤ 0.01, *** P ≤ 0.001, and **** P ≤ 0.0001).

Figure 3. ACT with NX-0255 treated Pmel-1 is associated with increased anti-tumor activity

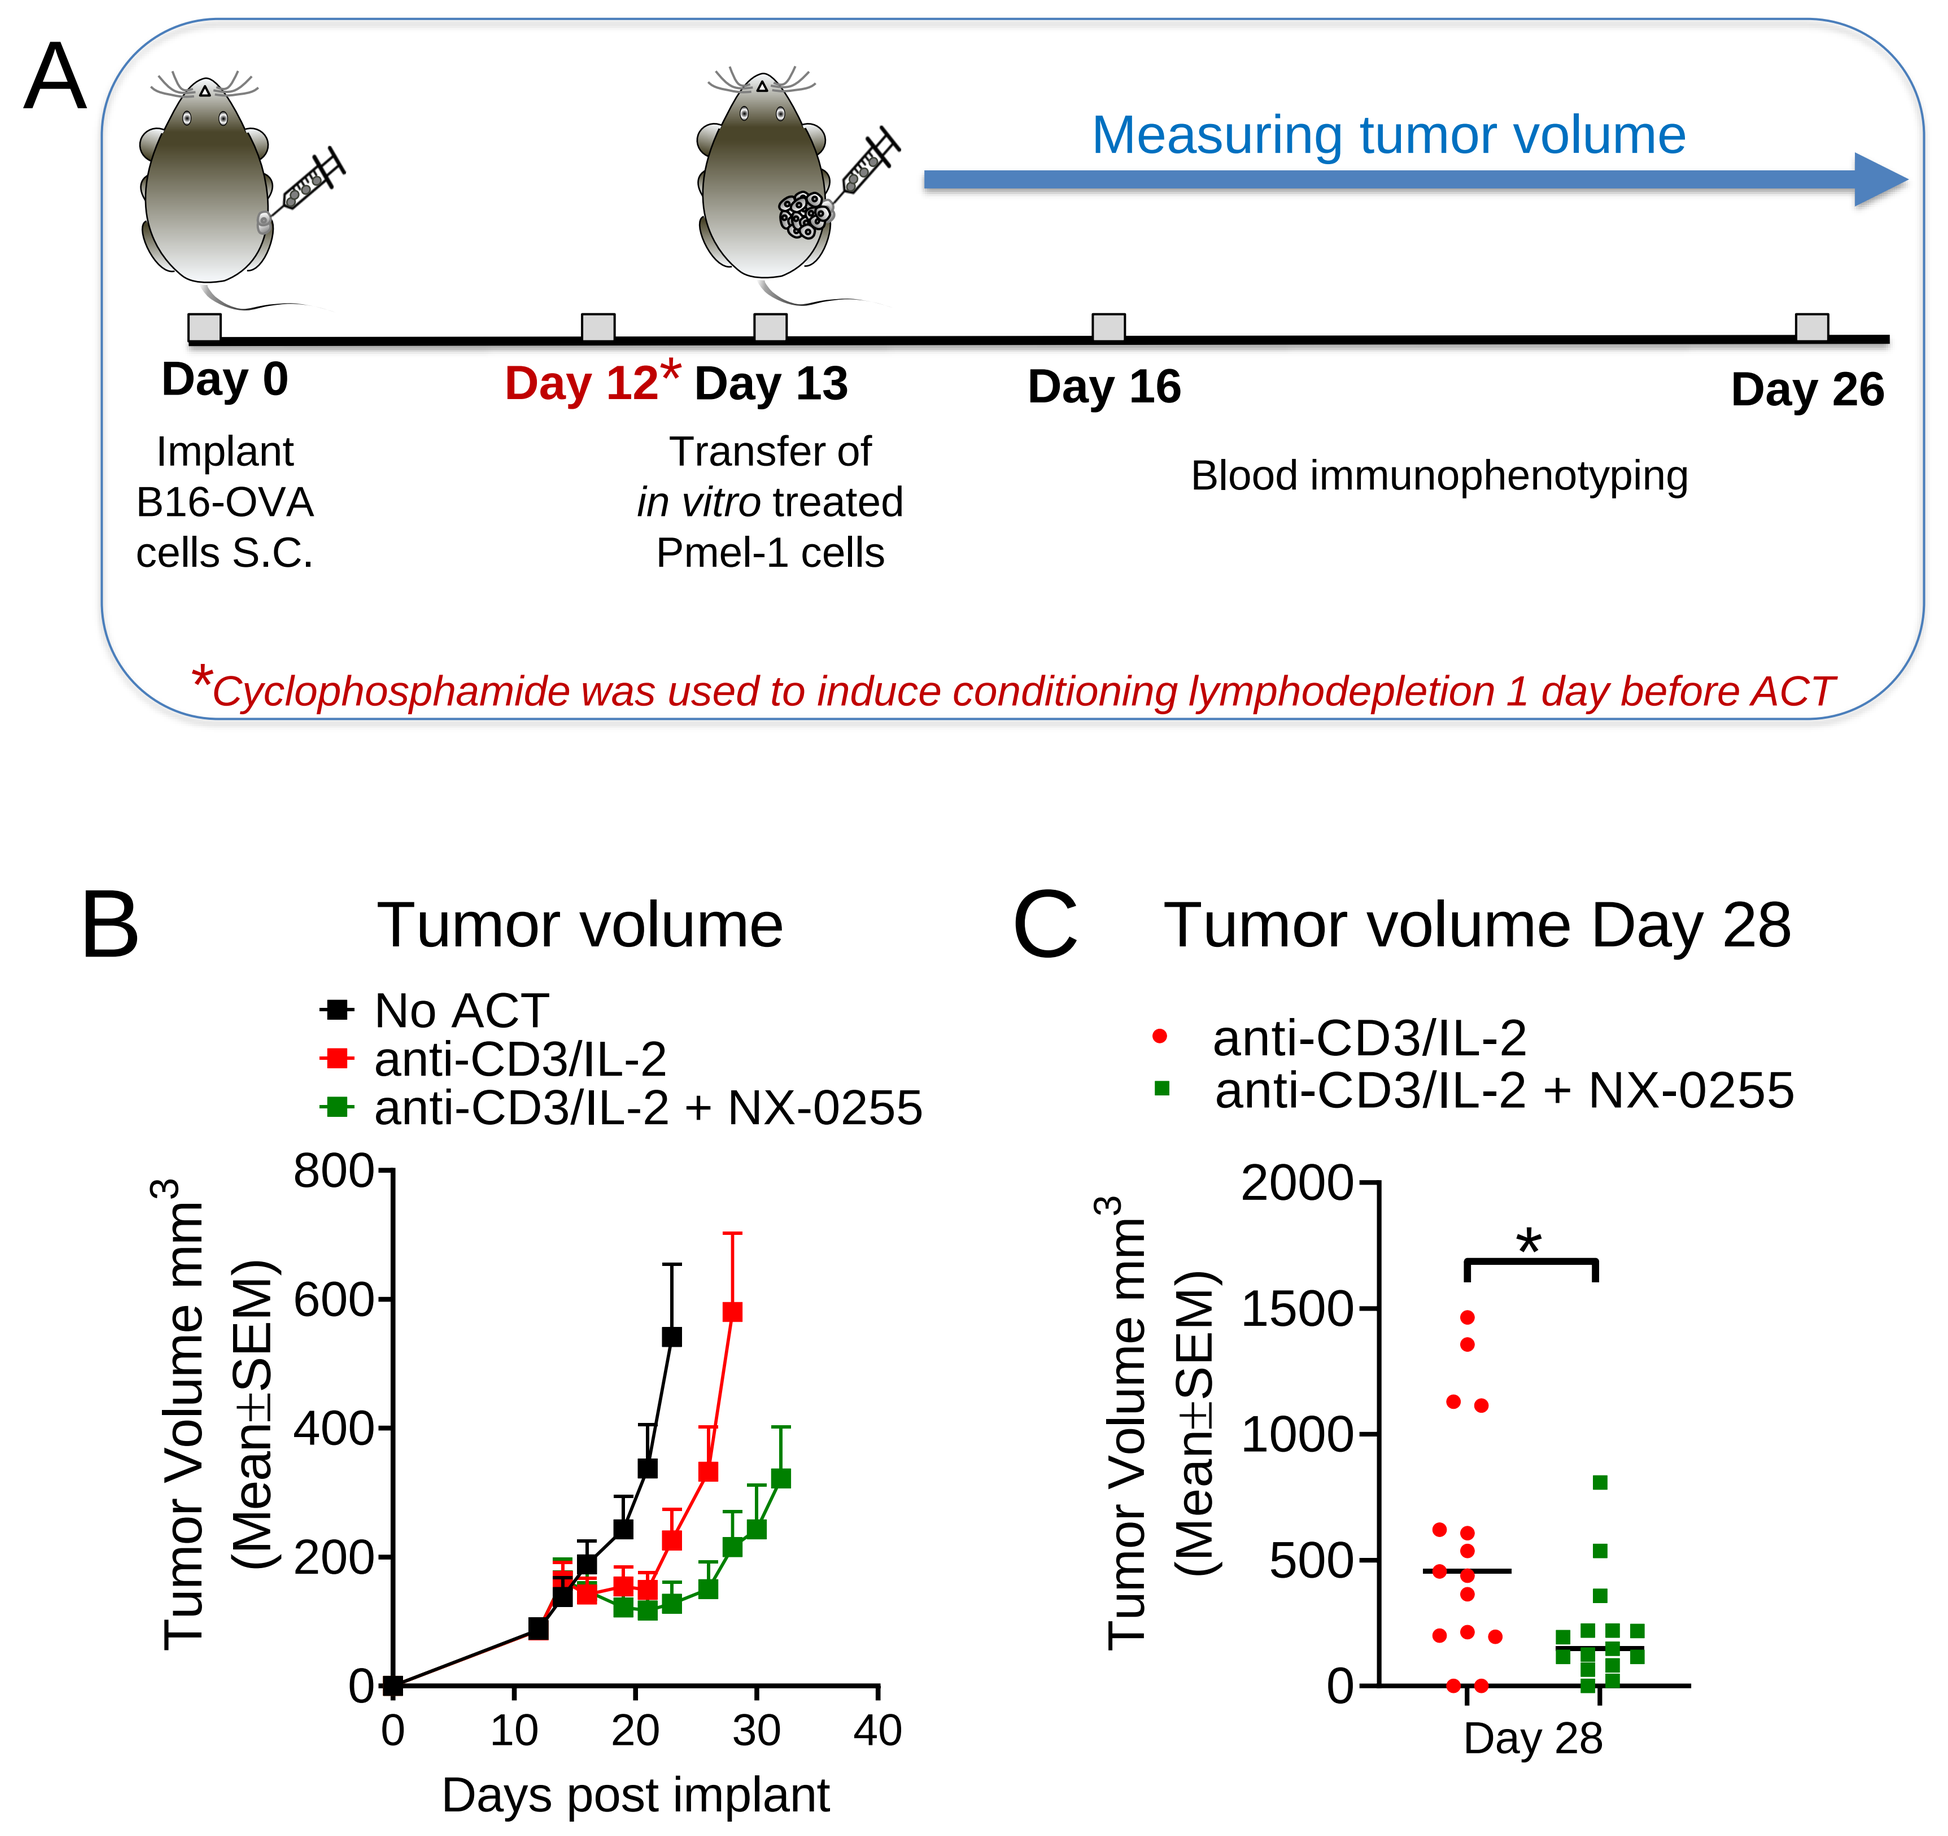
A
Measuring tumor volume
Day 0
Day 12*
Day 13
Day 16
Day 26
Implant B16-OVA cells S.C.
Transfer of *in vitro* treated Pmel-1 cells
Blood immunophenotyping
*Cyclophosphamide was used to induce conditioning lymphodepletion 1 day before ACT
B
Tumor volume
No ACT
anti-CD3/IL-2
anti-CD3/IL-2 + NX-0255
Tumor Volume mm3 (Mean±SEM)
Days post implant
C
Tumor volume Day 28
anti-CD3/IL-2
anti-CD3/IL-2 + NX-0255
Day 28

(A-D) Mice bearing B16-OVA s.c. tumors were treated IV on Day 13 with ACT of CD3-stimulated Pmel-1 T cells that were cultured in the presence of either 300 IU/mL IL-2 or 1 µM NX-0255 and 300 IU/mL IL-2. No ACT control mice were administered only media IV. Figure 3A shows a schematic of the study and sampling time points. Figure 3B shows mean tumor volumes ± SEM, on Days 0 through 32; lines end for each group at the last day that all animals were alive. Figure 3C shows individual tumor volumes with lines at group median values on Day 28, the last day that all the ACT treated animals were alive. Statistical significance of differences in mean tumor volumes between groups on Day 28 was evaluated using Mann-Whitney U test (P > 0.05, * P ≤ 0.05, ** P ≤ 0.01, *** P ≤ 0.001, and **** P ≤ 0.0001).

Figure 4. NX-0255 treated Pmel-1 cells show increased persistence and memory phenotype

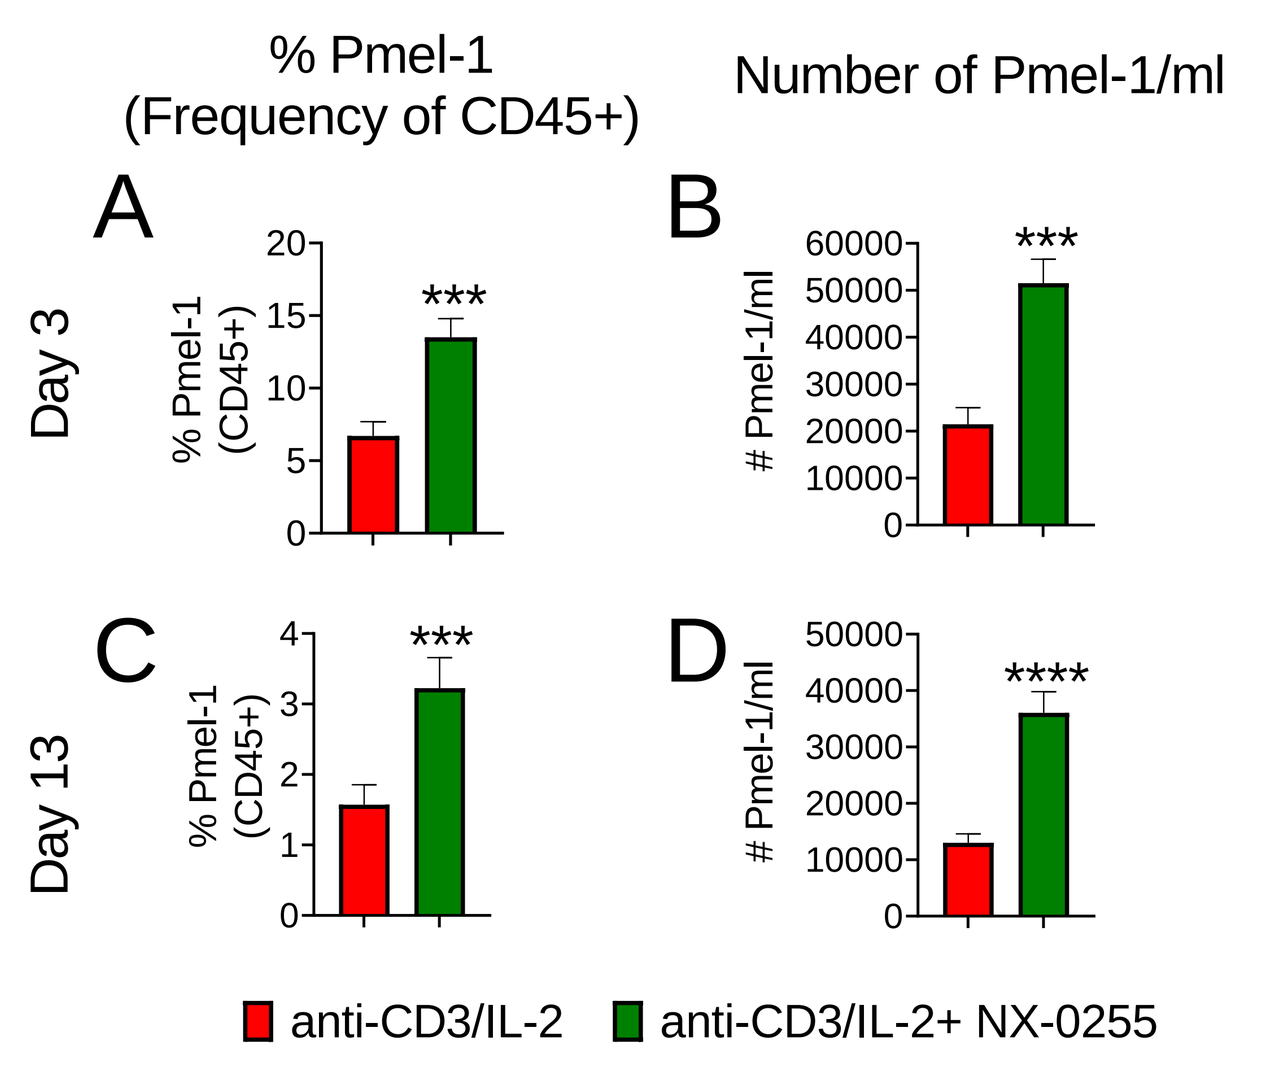
% Pmel-1
(Frequency of CD45+)
Number of Pmel-1/ml
Day 3
Day 13
anti-CD3/IL-2
anti-CD3/IL-2+ NX-0255

(A-L) Mice bearing B16-OVA s.c. tumors were treated IV on Day 13 with ACT of CD3-stimulated Pmel-1 T cells that were cultured in the presence of either 300 IU/mL IL-2 or 1 µM NX-0255 and 300 IU/mL IL-2. On Day 16 (3 days after ACT) and Day 26 (13 days after ACT) blood was collected from all ACT-treated animals and the frequency of Pmel-1 cells was determined by flow cytometry (A, Day 3; C, Day 13). The total number of Pmel-1 cells in the blood was assessed per mL of blood (B, Day 3; D, Day 13). The frequency of Pmel-1 cells with central-memory, effector-memory, memory-precursor and short-lived effector phenotype was determined at Day 3 (E, F, I and J) and at Day 13 (G, H, K and L) respectively. Group mean values ± SEM are shown in each graph. Statistical significance was determined by Mann-Whitney U test (*** P ≤ 0.001, and **** P ≤ 0.0001).

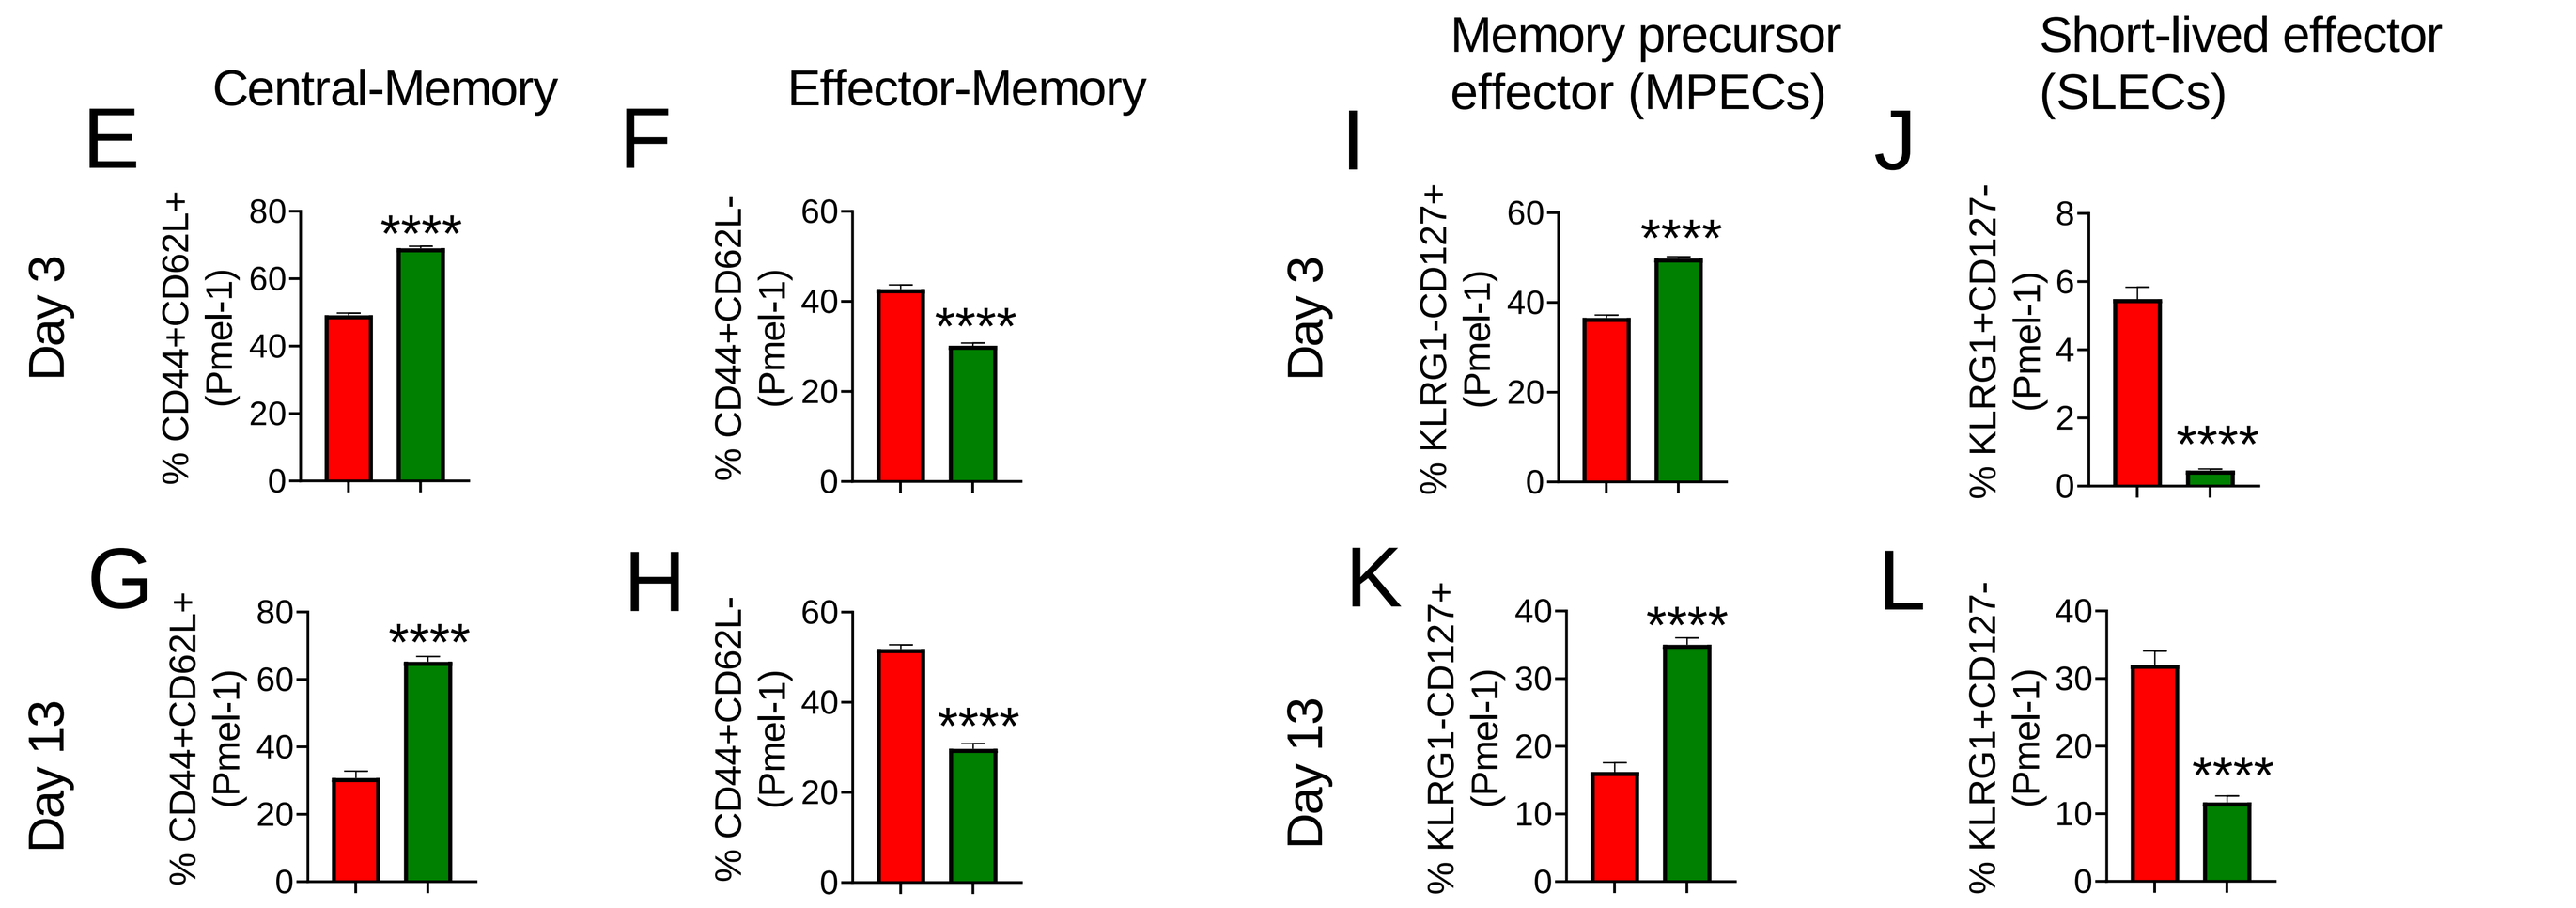
Central-Memory
Effector-Memory
Memory precursor effector (MPECs)
Short-lived effector (SLECs)
Day 3
Day 13
% CD44+CD62L+ (Pmel-1)
% CD44+CD62L- (Pmel-1)
% KLRG1-CD127+ (Pmel-1)
% KLRG1+CD127- (Pmel-1)

Figure 5. NX-0255 treated OT-I cells show increased persistence and rapid recall response upon re-challenge

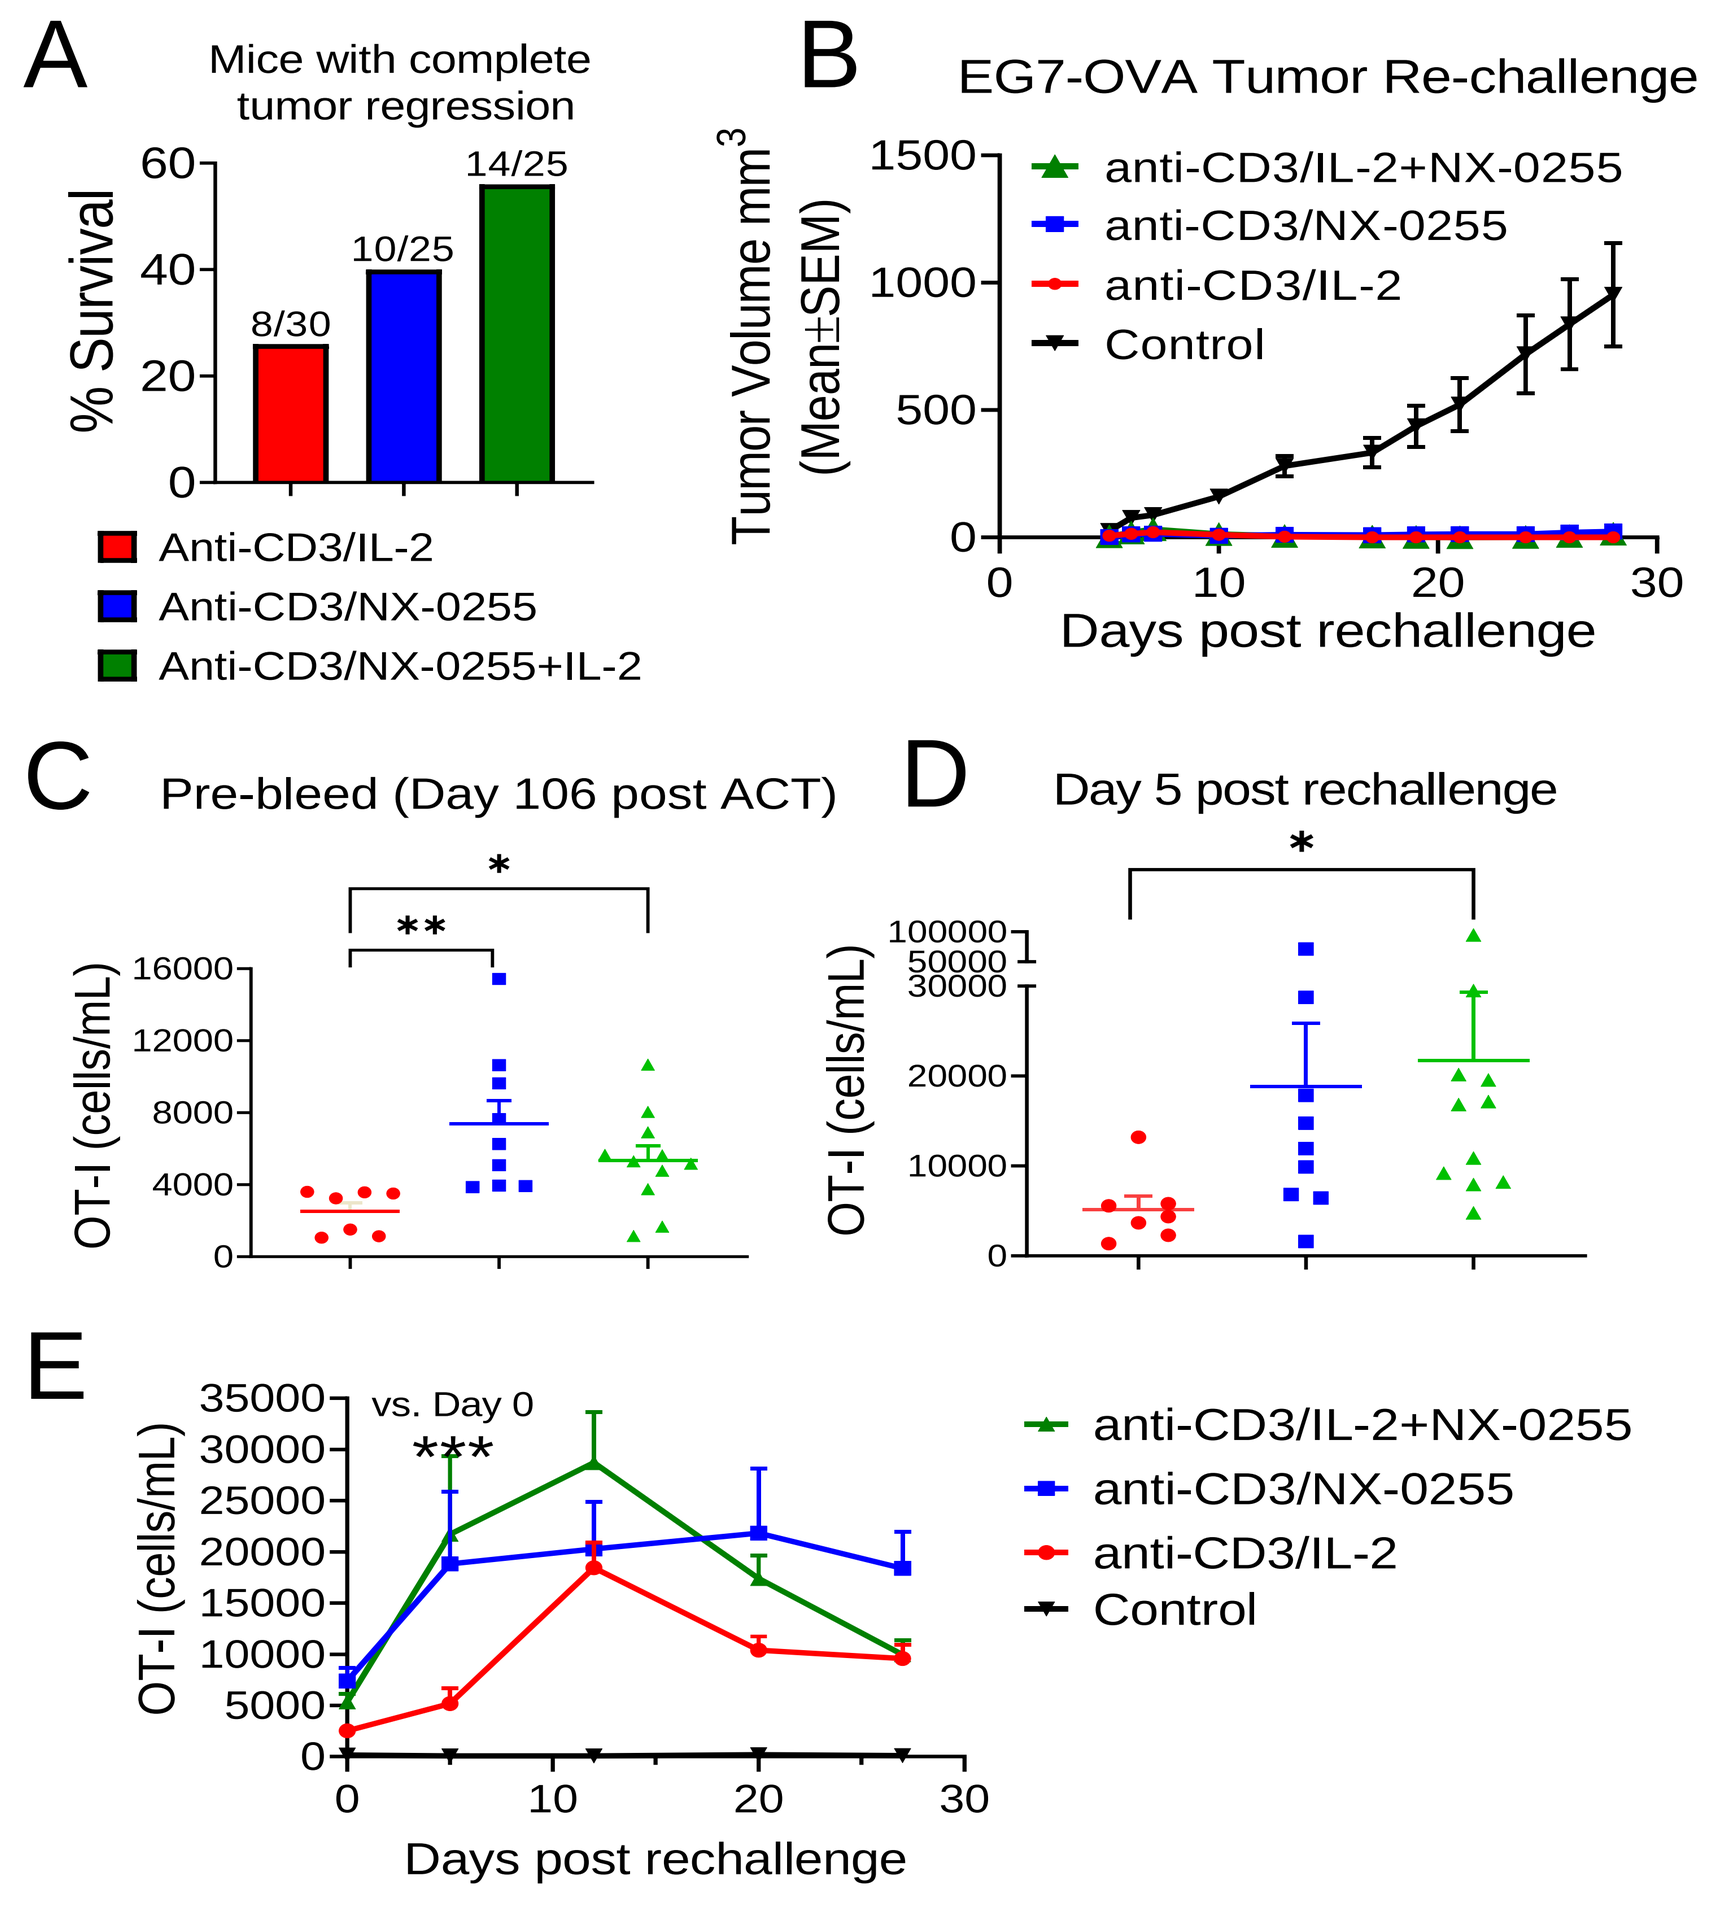
A
Mice with complete tumor regression
% Survival
8/30
10/25
14/25
Anti-CD3/IL-2
Anti-CD3/NX-0255
Anti-CD3/NX-0255+IL-2
B
EG7-OVA Tumor Re-challenge
anti-CD3/IL-2+NX-0255
anti-CD3/NX-0255
anti-CD3/IL-2
Control
Tumor Volume mm3 (Mean±SEM)
Days post rechallenge
C
Pre-bleed (Day 106 post ACT)
D
Day 5 post rechallenge
OT-I (cells/mL)
E
vs. Day 0
OT-I (cells/mL)
Days post rechallenge

(A-E) Mice treated with ACT of CD3-stimulated OT-I CD8+ T cells that were treated in the presence of either: 300 IU/mL IL-2, 1 µM NX-0255 or 1 µM NX-0255 and 300 IU/mL IL-2, and had complete tumor regression (A, percent survival from two studies) were re-challenged with E.G7-OVA cells 106 days after the initial ACT. "Control" group of mice were age-matched naïve mice, inoculated with E.G7-OVA cells. Tumor sizes were plotted as average tumor volume ± SEM (B). Persistence of adoptively transferred OT-I CD8+ T cells was assessed 106 days after the initial ACT. NX-0255 treated OT-I show increased persistence compared to IL-2 treated OT-I, with more than twice as many circulating OT-I present in the blood 106 days after the initial ACT (C; data from pre-bleed). The total number of OT-I cells in the blood was assessed per mL of total blood at Day 5 post rechallenge (D) and throughout the recall response until day 27 (E). Individual and mean values ± SEM are shown. Statistical significance was determined by one-way ANOVA test with Kruskal-Wallis multiple comparisons test (* P ≤ 0.05, ** P ≤ 0.01, *** P ≤ 0.001).

Figure 6. NX-0255+IL-2 treated OT-I cells differentiate into long-lived central-memory cells with high polyfunctionality and a broad range of TCR affinity

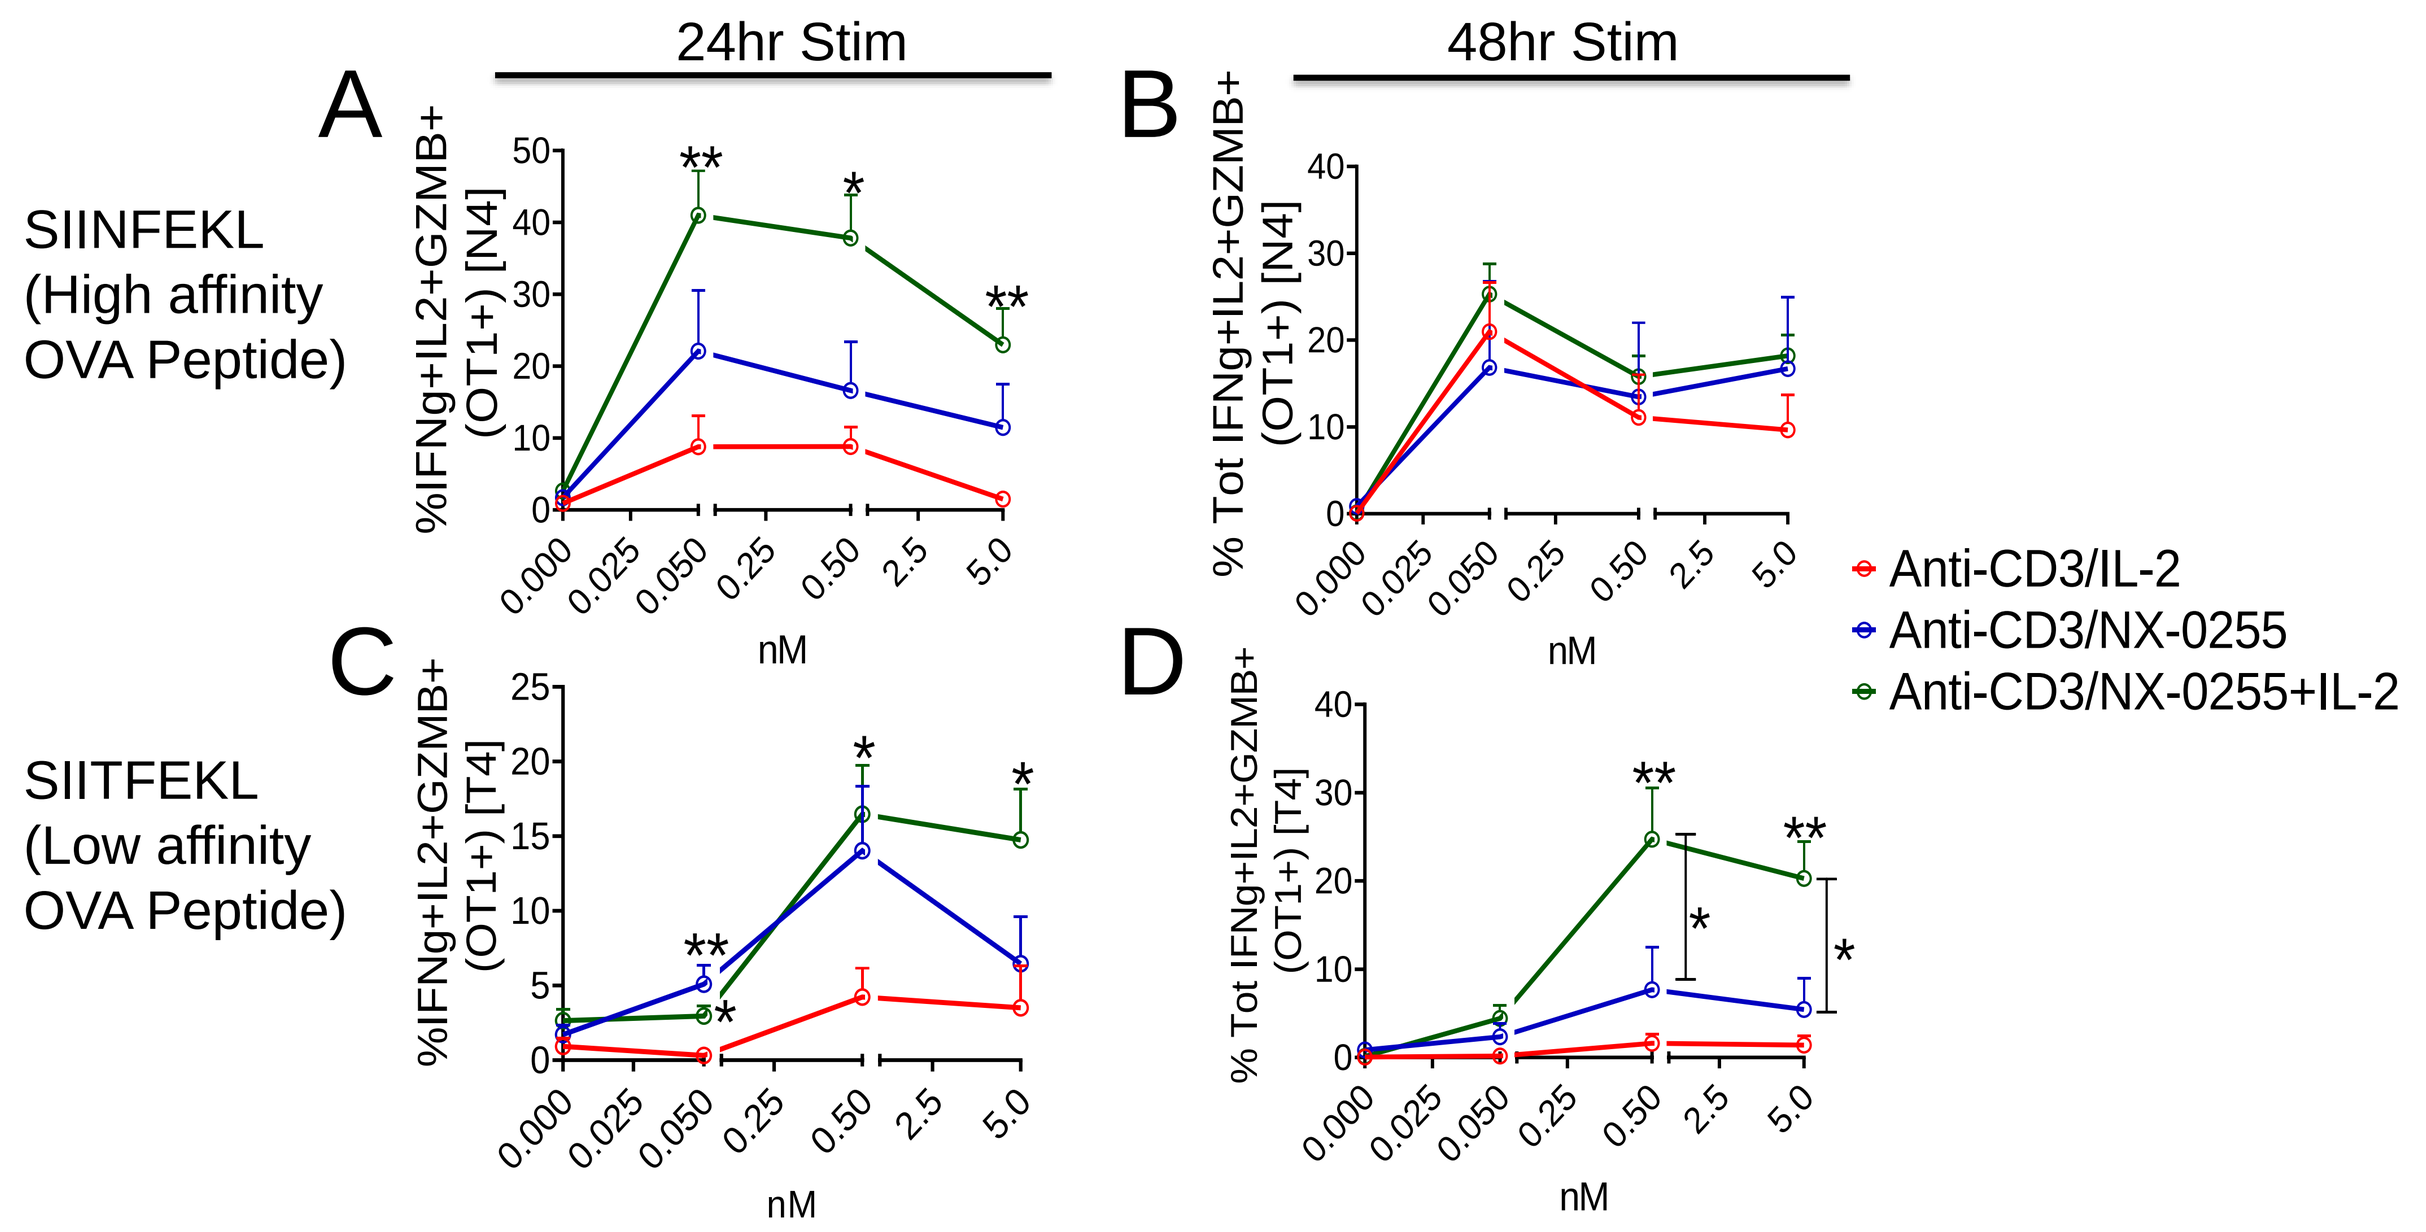
24hr Stim
48hr Stim
SIINFEKL
(High affinity OVA Peptide)
SIITFEKL
(Low affinity OVA Peptide)
%IFNg+IL2+GZMB+ (OT1+) [N4]
% Tot IFNg+IL2+GZMB+ (OT1+) [N4]
%IFNg+IL2+GZMB+ (OT1+) [T4]
% Tot IFNg+IL2+GZMB+ (OT1+) [T4]
nM
Anti-CD3/IL-2
Anti-CD3/NX-0255
Anti-CD3/NX-0255+IL-2

(A-D) Splenocytes from mice rechallenged with E.G7-OVA cells 106 days after the initial ACT were stimulated with high affinity (SIINFEKL - N4) and low affinity (SIITFEKL - T4) OVA peptides for 24 and 48 h in the presence of brefeldin A (added in the last 4 h); Antigen-specific OT-I CD8+ T cells were analyzed using flow cytometry intracellular staining and assessed for polyfunctionality (defined as co-expression of INF-g, IL-2 and Granzyme B). Curve graphs illustrate percentage of IFN-γ+IL-2+GranzB+ cells of total OT-I cells for each peptide concentration. NX-0255+IL-2 treated OT-I show increased percentage of polyfunctional cells compared to IL-2 treated OT-I, when stimulated with both high affinity (N4) and low affinity (T4) OVA peptides for 24 h (A and C). Similar percentage of polyfunctional OT-I cells was observed for all the groups when stimulated with high affinity (N4) for 48 h (B). NX-0255+IL-2 treated OT-I show increased percentage of polyfunctional cells compared to NX-0255 treated or IL-2 treated OT-I, when stimulated with low affinity (T4) OVA peptides for 48 h (D). Curve graphs display mean and SEM. Statistical significance was determined by one-way ANOVA test with Kruskal-Wallis multiple comparisons test (* P ≤ 0.05, ** P ≤ 0.01, *** P ≤ 0.001).

Conclusions

- Treatment of CD3-stimulated tumor-specific CD8 T cells using NX-0255, a novel small molecule CBL-B inhibitor, is associated with increased anti-tumor activity in both OT-I and Pmel-1 models.
- NX-0255 treated tumor-specific CD8+ T cells show increased expansion in tumor and blood, decreased surface expression of exhaustion markers and increased memory phenotype, after adoptive transfer *in vivo*.
- ACT with NX-0255 treated OT-I is associated with increased persistence and rapid recall response upon re-challenge.
- NX-0255+IL-2 treated OT-I cells differentiate into long-lived central-memory cells with superior polyfunctionality upon both high and low affinity antigen stimulation during the recall response.
- These results support the rationale for the use of NX-0255 in the production of an investigational drug-enhanced TIL therapy, DeTIL-0255, which is currently in a Phase 1 clinical trial [NCT05107739].

Disclosures

All authors are past or current employees of Nurix Therapeutics and hold company stock or stock options.

Presented at the American Association for Cancer Research (AACR) Annual Meeting. April 8-13, 2022. DO NOT POST